1244P
Annual Congress of the European Society for Medical Oncology (ESMO), Madrid, Spain September 8–12, 2017

Hospitalization Rates in COLUMBUS Part 1: A Phase 3 Trial of Encorafenib Plus Binimetinib vs Vemurafenib or Encorafenib Monotherapy in *BRAF*-Mutant Melanoma

Ana Arance,[1] Reinhard Dummer,[2] Paolo A. Ascierto,[3] Helen J. Gogas,[4] Mario Mandala,[5] Gabriella Liszkay,[6] Claus Garbe,[7] Dirk Schadendorf,[8] Ivana Krajsová,[9] Ralf Gutzmer,[10] Vanna Chiarion Sileni,[11] Caroline Dutriaux,[12] Jan Willem B. de Groot,[13] Naoya Yamazaki,[14] Carmen Loquai,[15] Laure A. de Parseval,[16] Michael D. Pickard,[17] Victor Sandor,[17] Caroline Robert,[18] Keith T. Flaherty[19]

[1]Hospital Clinic of Barcelona, Barcelona, Spain; [2]University Hospital Zürich Skin Cancer Center, Zürich, Switzerland; [3]Istituto Nazionale Tumori Fondazione Pascale, Naples, Italy; [4]University of Athens, Laikon Hospital, Athens, Greece; [5]Papa Giovanni XXIII Cancer Center Hospital, Bergamo, Italy; [6]National Institute of Oncology, Budapest, Hungary; [7]Eberhard Karls University, Tuebingen, Germany; [8]University Hospital Essen, Essen, Germany and German Cancer Consortium, Heidelberg, Germany; [9]University Hospital Prague and Charles University First Medical Faculty, Prague, Czech Republic; [10]Hannover Medical School, Hannover, Germany; [11]Oncology Institute of Veneto IRCCS, Padua, Italy; [12]Centre Hospitalier Universitaire de Bordeaux, Hôpital Saint-André, Bordeaux, France; [13]Isala, Zwolle, Netherlands; [14]National Cancer Center Hospital, Tokyo, Japan; [15]University Medical Center, Mainz, Germany; [16]Novartis Pharma AG, Basel, Switzerland; [17]Array BioPharma Inc., Boulder, CO, USA; [18]Institute Gustave Roussy, Villejuif, France; [19]Massachusetts General Hospital, Boston, MA, USA

INTRODUCTION

- *BRAF* (V600) mutations are common (35%–50%) in melanoma.[1,2]
- In patients with *BRAF*-mutant locally advanced or metastatic melanoma, combination therapy with a BRAF inhibitor (BRAFi) and a mitogen-activated protein kinase kinase inhibitor improves outcomes compared with BRAFi alone, and such combinations have become the standard of care.[3,4]
- Malignant melanoma is associated with significant costs and healthcare resource use.[5,6]
 - A substantial portion of healthcare costs are linked to treatment-related adverse events (AEs), and AEs requiring hospitalization are a key driver of costs.[5,6]
- The objective of this analysis was to evaluate resource utilization based on hospitalization data for patients with unresectable or metastatic *BRAF* V600 mutant melanoma enrolled in the COLUMBUS trial (ClinicalTrials.gov, NCT01909453; EudraCT, 2013-001176-38).

METHODS

Study Design

- COLUMBUS was a 2-part, phase 3, randomized, open-label study of combination therapy with encorafenib plus binimetinib vs monotherapy with vemurafenib or encorafenib conducted at 162 clinical sites in 28 countries **(Figure 1)**.

Figure 1. Study Design

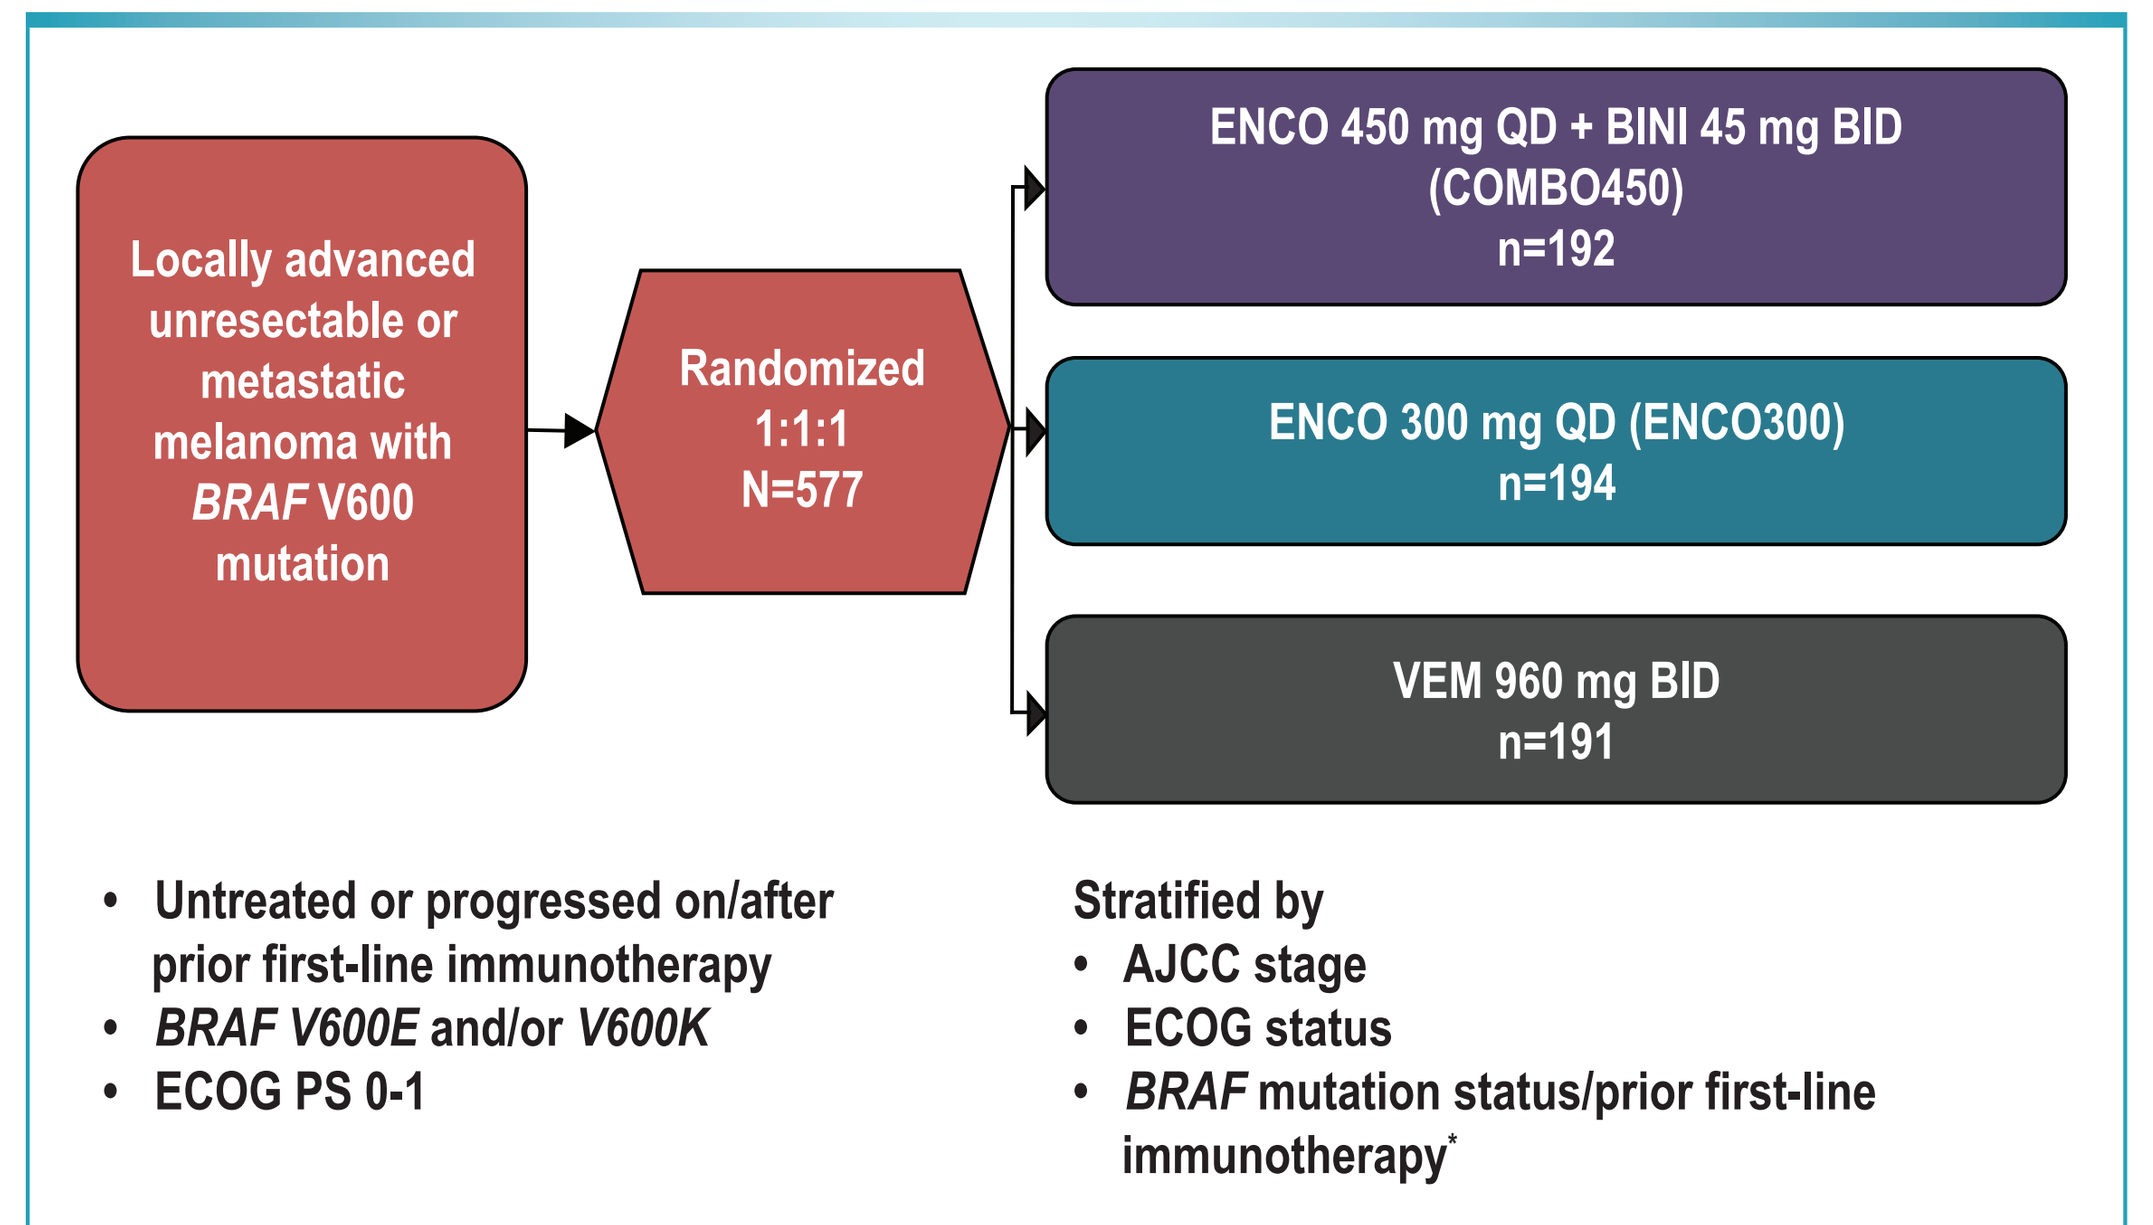
Locally advanced unresectable or metastatic melanoma with *BRAF* V600 mutation
Randomized 1:1:1 N=577
ENCO 450 mg QD + BINI 45 mg BID (COMBO450) n=192
ENCO 300 mg QD (ENCO300) n=194
VEM 960 mg BID n=191
• Untreated or progressed on/after prior first-line immunotherapy
• *BRAF V600E* and/or *V600K*
• ECOG PS 0-1
Stratified by
• AJCC stage
• ECOG status
• *BRAF* mutation status/prior first-line immunotherapy*

AJCC=American Joint Committee on Cancer; BID=twice daily; BINI=binimetinib; ECOG=Eastern Cooperative Oncology Group; ENCO=encorafenib; PS=performance status; QD=once daily; VEM=vemurafenib.
*Prior first-line immunotherapy replaced BRAF mutation status as a stratification factor after protocol amendment 2.

METHODS (continued)

- In Part 1, patients were randomized (1:1:1) to encorafenib 450 mg once daily plus binimetinib 45 mg twice daily (COMBO450), encorafenib 300 mg once daily (ENCO300), or vemurafenib 960 mg twice daily (VEM).

Key Inclusion Criteria

- Adults (age ≥18 years) with unresectable or metastatic *BRAF* V600E and/or V600K mutant melanoma
- Untreated or progressed on/after prior first-line immunotherapy for advanced or metastatic disease
- Eastern Cooperative Oncology Group performance status 0–1

Resource Utilization

- Information on hospitalizations that occurred during the study treatment period were characterized, including time to first occurrence (ie, time from the start of study drug to the date of first hospitalization), length of stay, and reason for hospital admission (ie, all causes, AE related).

Statistical Analysis

- The cutoff date for these analyses was May 19, 2016.
- Analyses were conducted in the safety population (all patients who received ≥1 dose of study drug and had ≥1 postbaseline safety evaluation) and included only hospitalizations that started on or after the first dose of study drug and up to 30 days after the last dose of study drug.
- Exposure-adjusted hospitalization rates (95% CIs) were calculated as the number of patients with a hospitalization divided by the total time they were at risk for their first hospitalization. Approximate 95% CIs were estimated by assuming a normal distribution.[7]
- Exposure-adjusted duration of hospitalization was calculated as the total duration that patients were hospitalized in each arm divided by the total duration of treatment with study drug.
- Time to onset of first hospitalization was determined by the Kaplan-Meier methodology.

RESULTS

Patients

- Among 577 patients enrolled, 192 were randomized to the COMBO450 arm, 194 to the ENCO300 arm, and 191 to the VEM arm; demographic and baseline characteristics were generally similar among treatment groups **(Table 1)**.

Table 1. Patient Demographics and Baseline Characteristics

Variable	COMBO450 (n=192)	ENCO300 (n=194)	VEM (n=191)
Mean (SD) age, y	56 (14)	55 (13)	55 (14)
Male sex, n (%)	115 (60)	108 (56)	111 (58)
ECOG PS, n (%)			
0	136 (71)	140 (72)	140 (73)
1	56 (29)	54 (28)	51 (27)
LDH normal or higher, %	55 (29)	47 (24)	52 (27)
≥3 organs involved, %	87 (45)	86 (44)	87 (46)
Previous immunotherapy, n (%)	57 (30)	58 (30)	57 (30)
Ipilimumab	7 (4)	10 (5)	7 (4)
Anti-PD1/PDL1	1 (1)	2 (1)	0

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ECOG=Eastern Cooperative Oncology Group; ENCO300=encorafenib 300 mg once daily; LDH=lactate dehydrogenase; PD1/PDL1=anti-programmed death-1/PD-ligand 1; PS=performance status; VEM=vemurafenib.

- The analyzed population comprised the entire safety set, which included 192 patients in the COMBO450 arm, 192 patients in the ENCO300 arm, and 186 patients in the VEM arm.
- Total duration of exposure to study drug was approximately 2400 patient-months in the COMBO450 arm, 1873 patient-months in the ENCO300 arm, and 1537 patient-months in the VEM arm.

Hospitalizations

- The numbers of patients hospitalized for the first time on study per 100 patient-months of exposure were 2.3 (95% CI, 1.6–3.0) in the COMBO450 arm and 3.5 (95% CI, 2.5–4.5) and 4.4 (95% CI, 3.2–5.6) in the ENCO300 and VEM arms, respectively **(Figure 2)**.
- The numbers of patients hospitalized for adverse events for the first time on study per 100 patient-months of exposure were 2.2 (95% CI, 1.6–2.9), 3.2 (95% CI, 2.2–4.1), and 4.0 (95% CI, 2.9–5.2) in the COMBO450, ENCO300, and VEM arms, respectively **(Figure 2)**.

Figure 2. Number of Patients With a First Hospitalization per 100 Patient-Months of Exposure to Study Drug

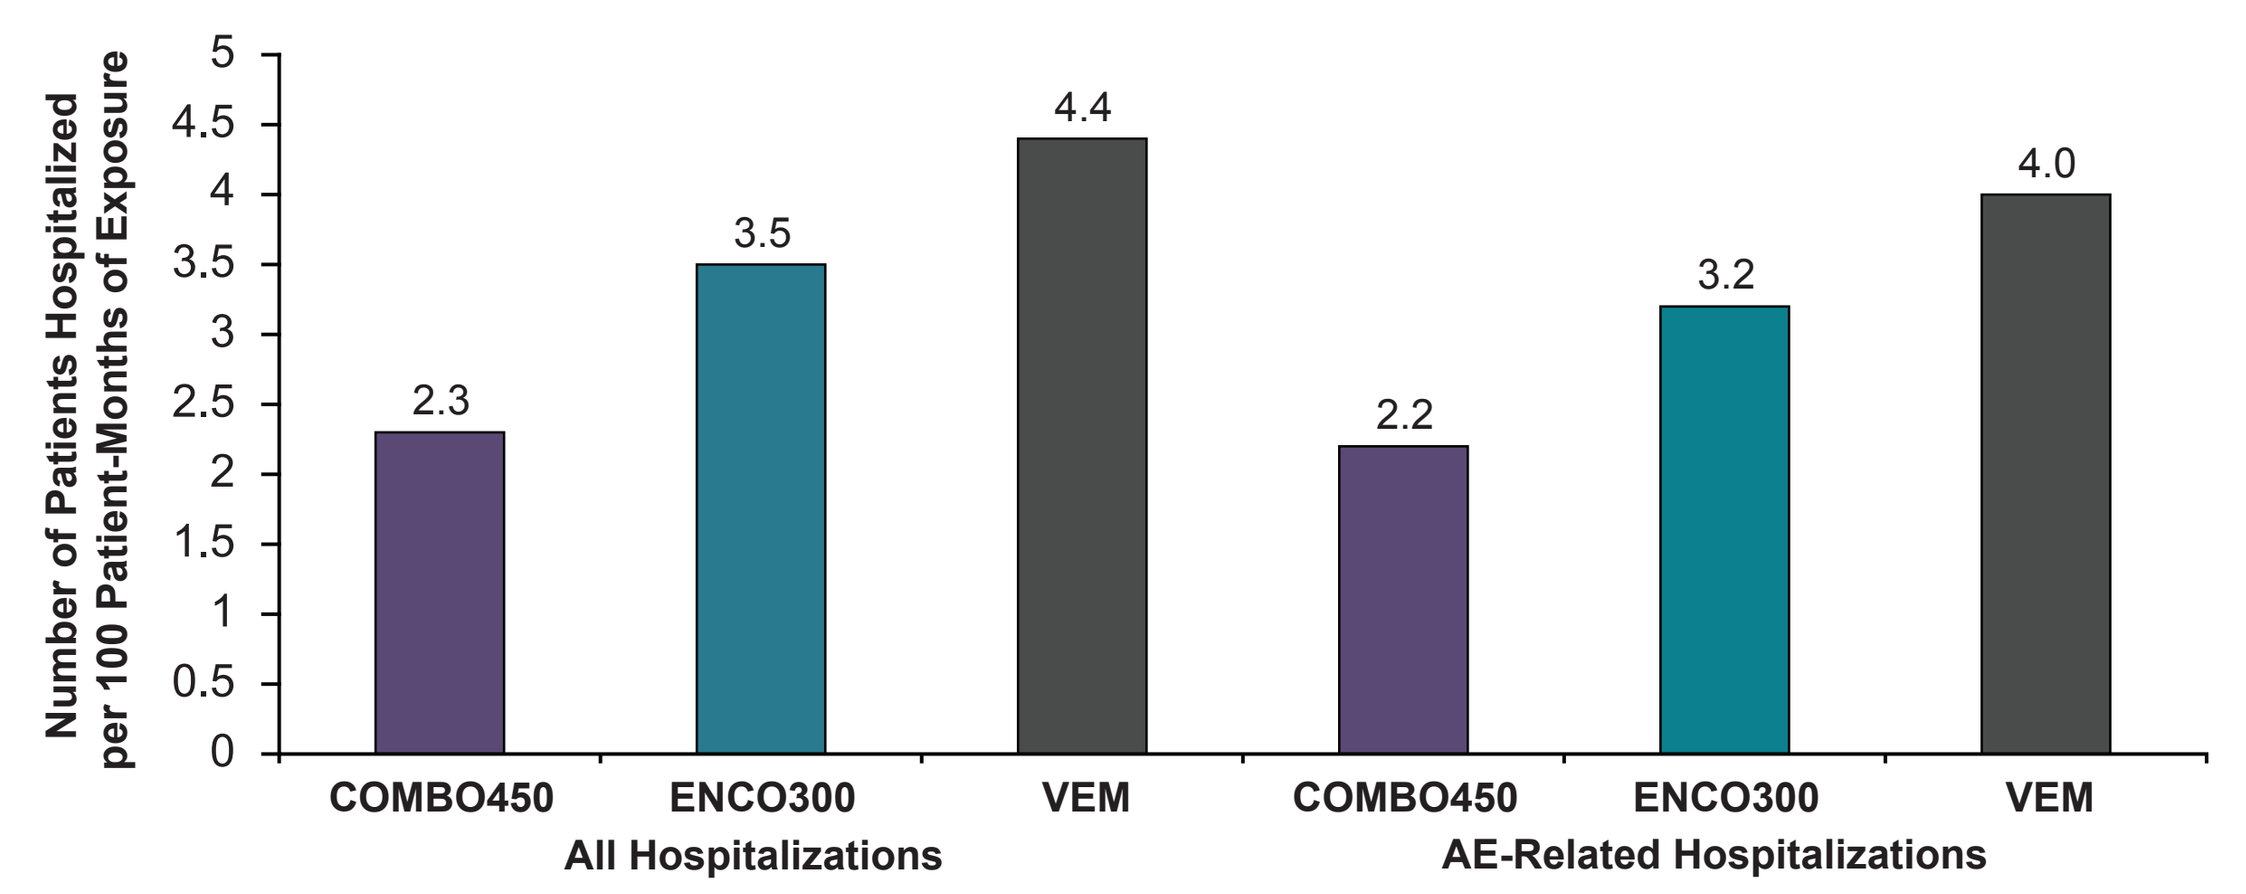
Number of Patients Hospitalized per 100 Patient-Months of Exposure
5
4.5
4
3.5
3
2.5
2
1.5
1
0.5
0
2.3
3.5
4.4
2.2
3.2
4.0
COMBO450
ENCO300
VEM
COMBO450
ENCO300
VEM
All Hospitalizations
AE-Related Hospitalizations

AE=adverse event; COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; VEM=vemurafenib.

Figure 3. Days of Hospitalization per 100 Patient-Months of Exposure

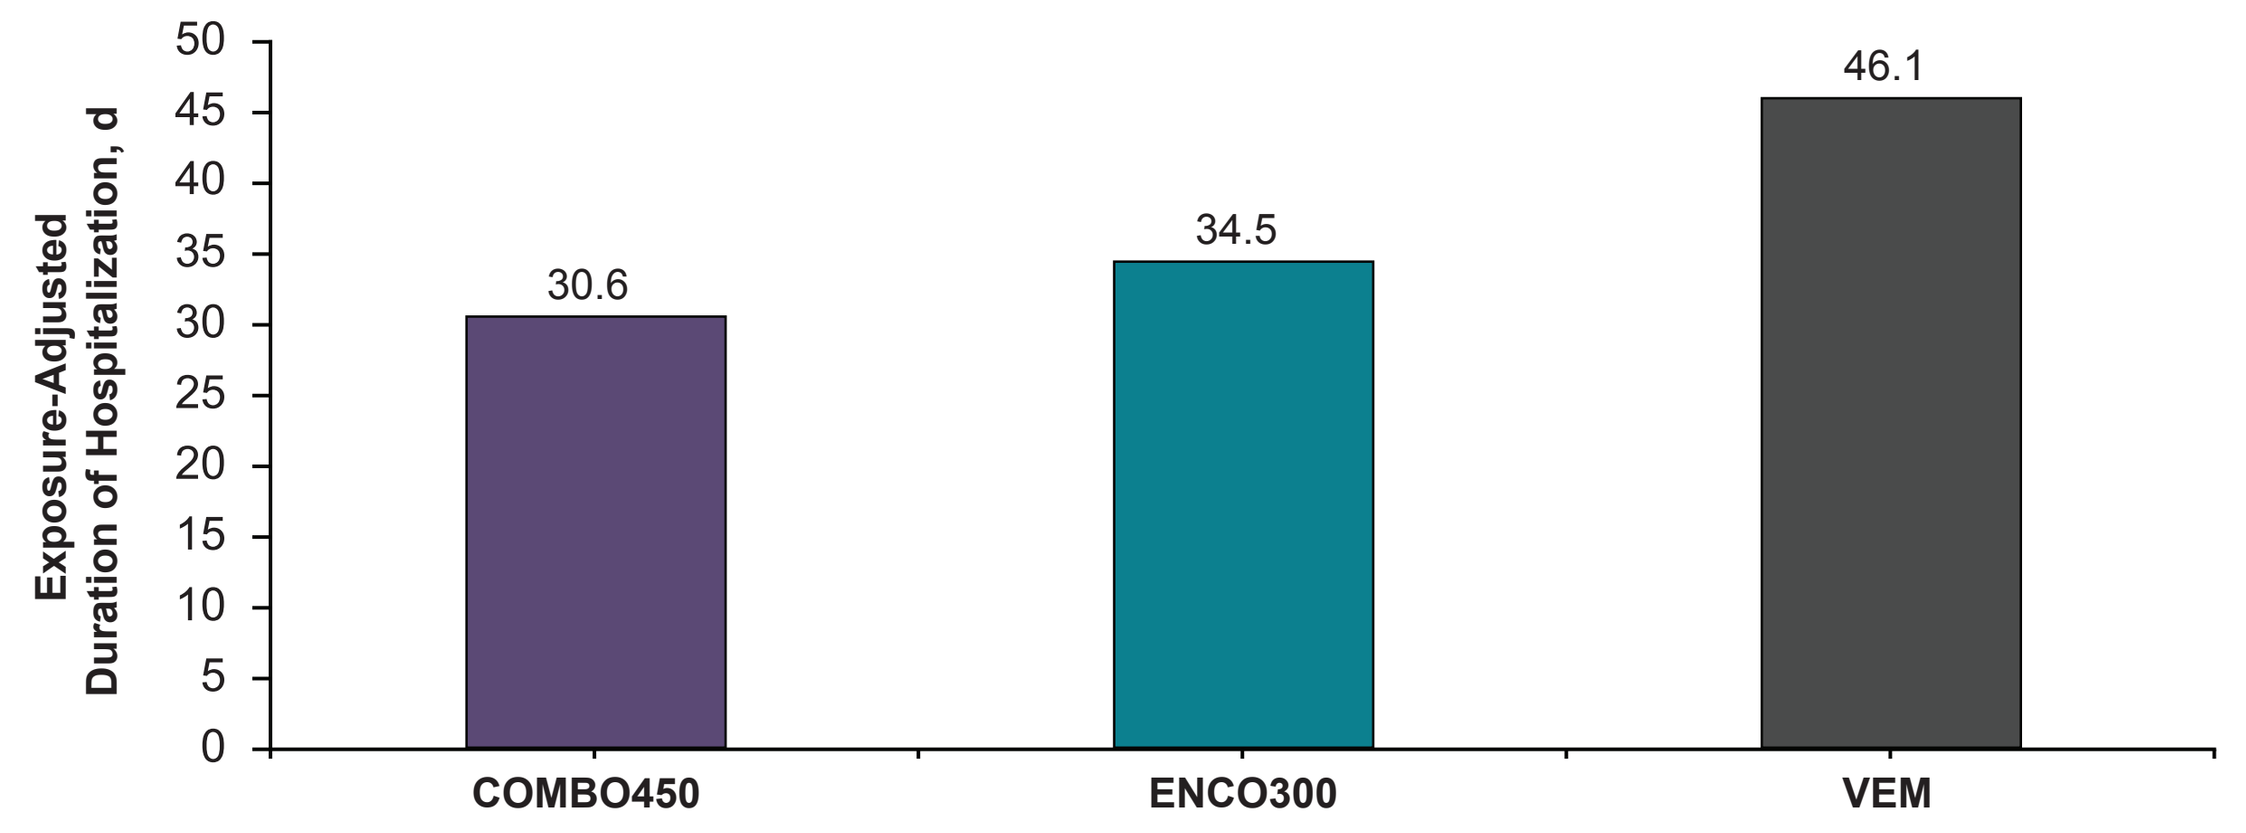
Exposure-Adjusted Duration of Hospitalization, d
50
45
40
35
30
25
20
15
10
5
0
30.6
34.5
46.1
COMBO450
ENCO300
VEM

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; VEM=vemurafenib.

- Patients were hospitalized for 30.6, 34.5, and 46.1 days per 100 patient-months of exposure in the COMBO450, ENCO300, and VEM arms, respectively **(Figure 3)**.
- Median (95% CI) time to first hospitalization for patients with ≥1 AE was longer in patients in the COMBO450 than in the ENC300 or VEM arms **(Table 2)**.

Table 2. Time to Onset of First Hospitalization

Treatment	Median Time to Onset (Months, 95% CI)
COMBO450	5.59 (2.99–6.64)
ENCO300	3.22 (1.91–4.60)
VEM	3.12 (2.20–4.37)

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; VEM=vemurafenib.

CONCLUSIONS

- In the COLUMBUS trial, total exposure to study drug was higher in the COMBO450 arm vs the ENCO300 or VEM monotherapy arms.
- After adjusting for exposure to study drug, COMBO450 was associated with fewer all-cause and AE-related hospitalization compared with ENCO300 or VEM.
- Moreover, the median time to first hospitalization was longer, and the exposure-adjusted length of stay was shorter, with COMBO450 arm compared with ENCO300 or VEM.

REFERENCES

1. Cancer Genome Atlas Network. *Cell.* 2015;161(7):1681-1696.
2. Krauthammer M, et al. *Nat Genet.* 2015;47(9):996-1002.
3. Daud A and Tsai K. *Oncologist.* 2017;22(7):823-833.
4. Ascierto PA, et al. *Lancet Oncol.* 2016;17(9):1248-1260.
5. Arondekar B, et al. *J Manag Care Spec Pharm.* 2015;21(2):158-164.
6. Bilir SP, et al. *Am Health Drug Benefits.* 2016;9(4):203-213.
7. He X, et al. J *Biom Biostat.* 2015;6:238.

DISCLOSURES

AA: Honoraria from and consulting/advisory role and speakers bureaus for Novartis, Roche, MSD, and BMS; travel expenses from Roche and BMS
RD: Honoraria from and consulting/advisory role for Roche, BMS, GSK, MSD, Novartis, and Amgen; research funding from Roche, BMS, GSK, MSD, and Novartis
PAA: Consulting fees from BMS, Roche/Genentech, MSD, Ventana, Novartis, Amgen, and Array BioPharma; research funding from BMS, Roche/Genentech, Ventana, and Array BioPharma
HJG: Consultant for Roche, BMS, MSD, Novartis, and Amgen
MM: Honoraria from Novartis, GSK, BMS, MSD, and Roche; speakers bureaus for Novartis, GSK, Roche, and BMS; advisory board member for Novartis, Amgen, MSD, and BMS; research funding from Roche
GL: Nothing to disclose
CG: Honoraria and travel expenses from and served in a consulting/advisory role and as speakers bureau member for Amgen, BMS, MSD, Novartis, Roche, and Philogen; has received research funding for University Hospital Tuebingen from BMS, Novartis, and Roche
DS: Honoraria and travel expenses from and consulting/advisory role and speakers bureaus for Amgen, BMS, Novartis, Roche, and MSD; research funding for University Hospital Essen from Amgen, BMS, Novartis, Roche, and MSD
IK: Advisory board member for BMS, Novartis, Roche, MSD; travel expenses from BMS and MSD
RG: Consulting fees and/or honoraria from Roche, BMS, MSD, GSK, Novartis, Almirall, LEO, Amgen, Pfizer, Pierre Fabre, Merck Serono, Boehringer Ingelheim; research funding from Roche, Novartis, Pfizer, Johnson & Johnson; travel expenses from BMS, Roche
VCS: Honoraria received from Novartis, GSK, BMS, MSD, and Roche; speakers bureaus for Novartis, GSK, Roche, and BMS; advisory board member for Novartis, Amgen, MSD, BMS, and Roche
CD: Nothing to disclose
JWBG: Consulting/advisory role for Amgen, Bayer, Celgene, Roche, BMS, GSK, MSD, and Merck Serono
NY: Advisory role for Chugai Pharma, Bristol-Myers Squibb Japan, and Ono Pharmaceutical; honoraria from Chugai Pharma, Bristol-Myers Squibb Japan, Ono Pharmaceutical, GSK, Takeda, AstraZeneca Japan, Boehringer Ingelheim, and Maruho
CL Advisory board member for Roche, Novartis, BMS, MSD, BioNTech, Pierre Fabre, and Amgen; speakers fees from Roche, Novartis, BMS, and MSD; travel expenses from Roche, Novartis, BMS, MSD, and Amgen
LP: Employee of Novartis Pharma AG; may own stock and/or stock options
MP: Employee of Array BioPharma; may own stock and/or stock options
VS: Employee/leadership role at Array BioPharma; stock and/or other ownership of Array BioPharma and Incyte Corp
CR: Consultant for Roche, Novartis, BMS, MSD, and Amgen
KF: Honoraria from and consulting/advisory role for Novartis and Array BioPharma; research funding from Novartis

Acknowledgments

Editorial support for development of this manuscript was provided by Bret Fulton at Complete Healthcare Communications, LLC (West Chester, PA), a CHC Group company, and funded by Array BioPharma Inc.

This study is sponsored by Array BioPharma Inc.

Available at:
www.arraybiopharma.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.