1245P

Annual Congress of the European Society for Medical Oncology (ESMO), Madrid, Spain
September 8–12, 2017

Quality of Life in COLUMBUS Part 1: A Phase 3 Trial of Encorafenib Plus Binimetinib vs Vemurafenib or Encorafenib Monotherapy in *BRAF*-Mutant Melanoma

Helen J. Gogas,[1] Reinhard Dummer,[2] Paolo A. Ascierto,[3] Ana Arance,[4] Mario Mandala,[5] Gabriella Liszkay,[6] Claus Garbe,[7] Dirk Schadendorf,[8] Ivana Krajsová,[9] Ralf Gutzmer,[10] Vanna Chiarion Sileni,[11] Caroline Dutriaux,[12] Jan Willem B. de Groot,[13] Naoya Yamazaki,[14] Carmen Loquai,[15] Laure A. de Parseval,[16] Michael Pickard,[17] Victor Sandor,[17] Caroline Robert,[18] Keith T. Flaherty[19]

[1]National and Kapodistrian University of Athens, Laikon Hospital, Athens, Greece; [2]University Hospital Zürich Skin Cancer Center, Zürich, Switzerland; [3]Istituto Nazionale Tumori Fondazione Pascale, Naples, Italy; [4]Hospital Clinic of Barcelona, Barcelona, Spain; [5]Papa Giovanni XXIII Cancer Center Hospital, Bergamo, Italy; [6]National Institute of Oncology, Budapest, Hungary; [7]Eberhard Karls University, Tuebingen, Germany; [8]University Hospital Essen, Essen, Germany, and German Cancer Consortium, Heidelberg, Germany; [9]University Hospital Prague and Charles University First Medical Faculty, Prague, Czech Republic; [10]Hannover Medical School, Hannover, Germany; [11]Oncology Institute of Veneto IRCCS, Padua, Italy; [12]Centre Hospitalier Universitaire de Bordeaux, Hôpital Saint-André, Bordeaux, France; [13]Isala, Zwolle, Netherlands; [14]National Cancer Center Hospital, Tokyo, Japan; [15]University Medical Center, Mainz, Germany; [16]Novartis Pharma AG, Basel, Switzerland; [17]Array BioPharma Inc., Boulder, CO, USA; [18]Institute Gustave Roussy, Villejuif, France; [19]Massachusetts General Hospital, Boston, MA, USA

INTRODUCTION

- Combination therapy with a BRAF inhibitor (BRAFi) and a mitogen-activated protein kinase kinase inhibitor (MEKi) is considered the standard of care in patients with *BRAF*-mutant locally advanced or metastatic melanoma, providing greater inhibition of the mitogen-activated protein kinase (MAPK) pathway and improved clinical efficacy compared with BRAFi monotherapy.[1]
- Preservation of health-related quality of life (HRQoL) is an important consideration of BRAFi/MEKi treatment.[2]
 - Results from phase 3 trials with other approved BRAFi/MEKi combinations showed that dabrafenib plus trametinib improved HRQoL compared with dabrafenib or vemurafenib alone,[2,3] whereas no difference in HRQoL was noted with vemurafenib plus cobimetinib vs vemurafenib alone.[4]
- The phase 3 COLUMBUS trial assessed a novel BRAFi/MEKi combination, encorafenib plus binimetinib, vs BRAFi monotherapy with encorafenib or vemurafenib.[5]
 - The study met its primary endpoint, demonstrating significantly longer progression-free survival with encorafenib plus binimetinib vs vemurafenib (14.9 vs 7.3 months; hazard ratio [HR], 0.54 [95% CI, 0.41–0.71]; *P*<0.001).
 - The tolerability profile of the combination was favorable compared with either vemurafenib or encorafenib monotherapy.
- The objective of this analysis was to compare patient-reported HRQoL between treatment arms in the COLUMBUS trial (ClinicalTrials.gov, NCT01909453; EudraCT, 2013-001176-38).

METHODS

Study Design

- COLUMBUS was a 2-part, phase 3, randomized, open-label, multicenter study of combination therapy with encorafenib plus binimetinib vs monotherapy with vemurafenib or encorafenib **(Figure 1)**.
- The study was conducted at 162 clinical sites in 28 countries.
- In part 1, patients were randomized (1:1:1) to encorafenib 450 mg once daily plus binimetinib 45 mg twice daily (COMBO450), encorafenib 300 mg once daily (ENCO300), or vemurafenib 960 mg twice daily (VEM).

Figure 1. Study Design for COLUMBUS Part 1

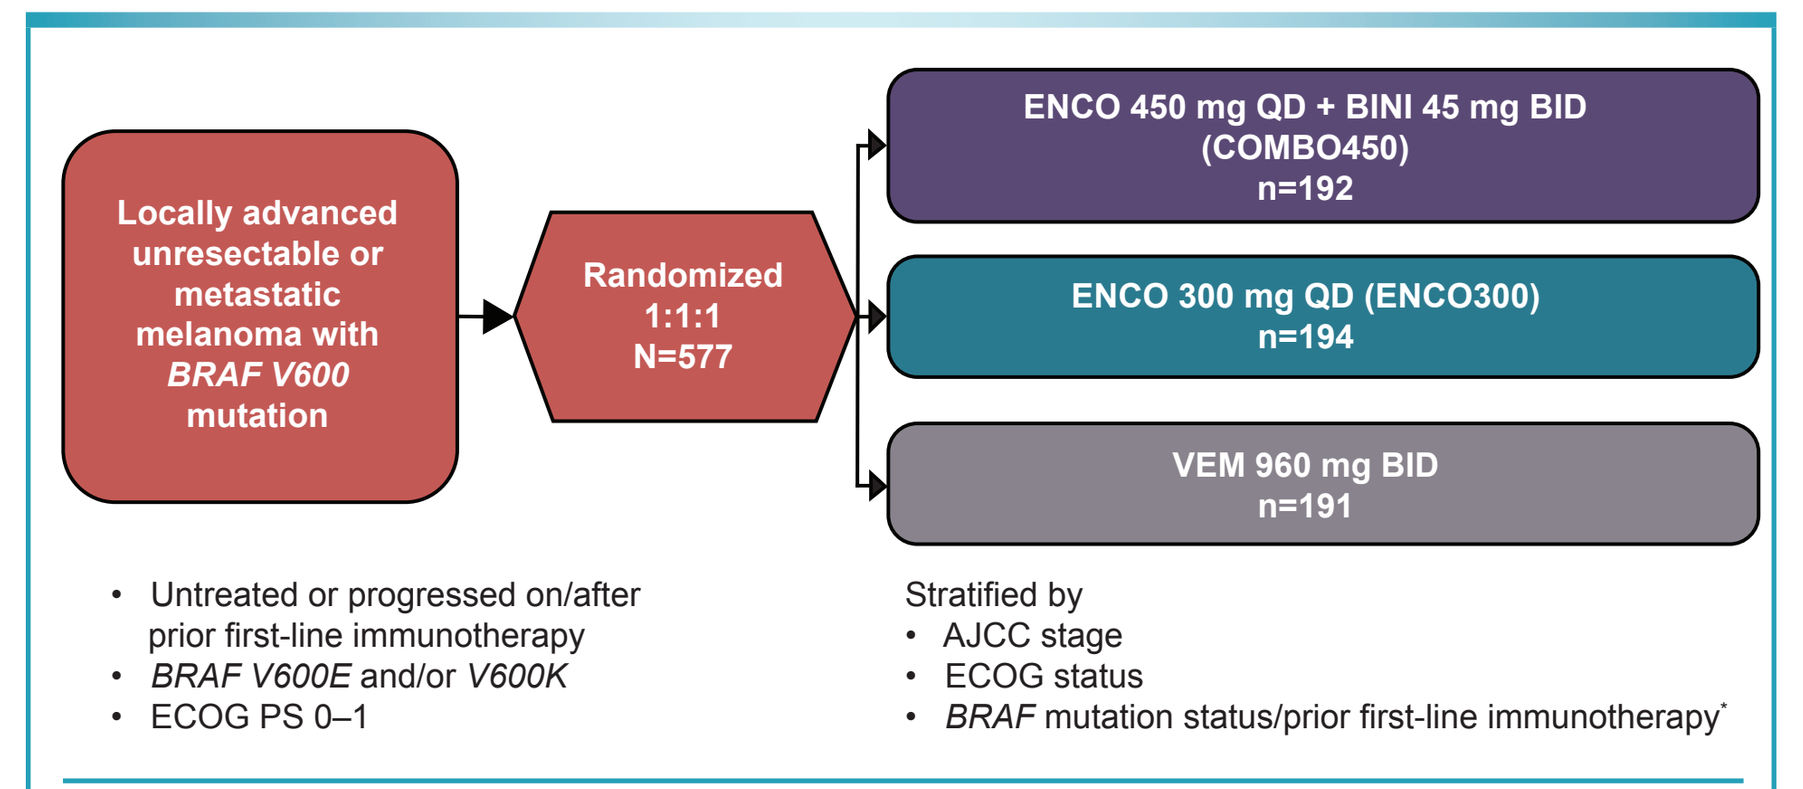
Locally advanced unresectable or metastatic melanoma with BRAF V600 mutation
Randomized 1:1:1 N=577
ENCO 450 mg QD + BINI 45 mg BID (COMBO450) n=192
ENCO 300 mg QD (ENCO300) n=194
VEM 960 mg BID n=191
• Untreated or progressed on/after prior first-line immunotherapy
• BRAF V600E and/or V600K
• ECOG PS 0–1
Stratified by
• AJCC stage
• ECOG status
• BRAF mutation status/prior first-line immunotherapy*

AJCC=American Joint Committee on Cancer; BID=twice daily; BINI=binimetinib; ECOG PS=Eastern Cooperative Oncology Group performance status; ENCO=encorafenib; QD=once daily; VEM=vemurafenib.
*Prior first-line immunotherapy replaced *BRAF* mutation status as a stratification factor after protocol amendment 2.

Key Inclusion Criteria

- Aged ≥18 years
- Unresectable or metastatic *BRAF* V600E or V600K mutant melanoma
- Untreated or progressed on or after prior first-line immunotherapy
- Eastern Cooperative Oncology Group performance status 0–1

Patient-Reported Outcomes

- Assessed at screening, every 8 weeks after randomization for the first 24 months and then every 12 weeks thereafter until disease progression, at end of treatment or end of study for patients who discontinued treatment for reasons other than progression, and at the safety follow-up visit
- Functional Assessment of Cancer Therapy–Melanoma (FACT-M, version 4)
 - The melanoma subscale, comprising 16 items related to signs, symptoms, and physical/social activities most relevant to patients with advanced-stage melanoma
 - Other items assessed in this study addressed physical well-being (7 items), functional well-being (7 items), social/family well-being (7 items), and emotional well-being (6 items).
 - Items were scored on a Likert scale representing degrees of frequency or difficulty ranging from 0 (not at all) to 4 (very much).
- European Organisation for Research and Treatment of Cancer core quality-of-life Questionnaire (EORTC QLQ-C30, version 3.0)[6]
 - 30-item questionnaire
 - 5 multi-item functional scales: physical, role, emotional, cognitive, social functioning; 3 multi-item symptom scales: fatigue, nausea/vomiting, pain
 - 6 single-item symptoms: dyspnea, insomnia, appetite loss, constipation, diarrhea, financial impact
 - Global health status scale
 - All scores for scales and single-item measures range from 0 to 100.

METHODS (continued)

Analyses

- HRQoL was assessed in the intention-to-treat population.
- The melanoma subscale of the FACT-M and the EORTC QLQ-C30 global health status scale were the primary patient-reported outcomes of interest for the study.
- Descriptive statistics were used to summarize the FACT-M melanoma subscale and EORTC QLQ-C30 global health status scores, using means and standard errors of the mean.
 - A post hoc exploratory analysis evaluated changes from baseline by baseline FACT-M and EORTC QLQ-C30 global health status scores with patients within each treatment arm divided into tertiles.
- Time to definitive 10% deterioration on the FACT-M melanoma subscale and EORTC QLQ-C30 global health status scale were analyzed using the Kaplan-Meier method.
 - Cox regression models, stratified by randomization stratification factors, were used to estimate the HRs and 2-sided 95% CI.
- A post hoc exploratory analysis evaluated changes from baseline in FACT-M melanoma subscale and EORTC QLQ-C30 global health status scores by baseline lactate dehydrogenase (LDH) status. A high LDH level is a strong independent predictor of shorter survival among patients with unresectable, metastatic melanoma.[7]
 - Mean changes in scores from baseline on the FACT-M melanoma subscale and EORTC QLQ-C30 global health status scale were calculated for patients within each arm stratified by baseline LDH levels of normal vs high.

RESULTS

Patients

- Among 577 patients randomized in Part 1 of the study, 192 were randomized to COMBO450, 191 were randomized to VEM, and 194 were randomized to ENCO300.
- Demographic and baseline characteristics are summarized in **Table 1**.
 - Characteristics were generally similar among treatment groups, including mean baseline HRQoL scores on the FACT-M melanoma subscale and EORTC QLQ-C30 global status scale.

Table 1. Patient Demographics and Baseline Characteristics

Variable	COMBO450 (n=192)	ENCO300 (n=194)	VEM (n=191)
Age, median (range), y	57 (20–89)	54 (23–88)	56 (21–82)
Male, n (%)	115 (60)	108 (56)	111 (58)
ECOG PS, n (%)			
0	136 (71)	140 (72)	140 (73)
1	56 (29)	54 (28)	51 (27)
LDH ≥ normal, %	55 (29)	47 (24)	52 (27)
≥3 organs involved, %	87 (45)	86 (44)	87 (46)
Baseline HRQoL score, mean (SEM)			
FACT- M melanoma subscale	52 (0.65)	53 (0.57)	52 (0.65)
EORT C QLQ-C30 global health status	67 (1.6)	66 (1.5)	65 (1.7)

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ECOG PS=Eastern Cooperative Oncology Group performance status; ENCO300=encorafenib 300 mg once daily; EORTC QLQ-C30=European Organisation for Research and Treatment of Cancer core quality-of-life questionnaire; FACT-M=Functional Assessment of Cancer Therapy–Melanoma; HRQoL=health-related quality of life; LDH=lactate dehydrogenase; VEM=vemurafenib.

- Compliance with completing HRQoL assessments was equivalent among the 3 treatment arms, ranging from 82–100% for the FACT-M **(Table 2)** and 82–96% for the EORTC QLQ-C30 **(Table 3)**.

Table 2. Patient Compliance on FACT-M Through Cycle 25

Visit	COMBO450 (n=192)		ENCO300 (n=194)		VEM (n=191)	
	Patients Remaining in the Study (n)	Patients Who Filled Out the Instrument (%)*	Patients Remaining in the Study (n)	Patients Who Filled Out the Instrument (%)*	Patients Remaining in the Study (n)	Patients Who Filled Out the Instrument (%)*
Baseline	192	86	194	82	191	84
Cycle 3	186	90	186	90	182	88
Cycle 5	174	88	158	92	146	88
Cycle 7	161	89	132	91	110	92
Cycle 9	139	88	100	88	87	92
Cycle 11	121	90	86	92	63	89
Cycle 13	104	90	77	92	50	96
Cycle 15	94	88	72	86	40	90
Cycle 17	83	92	62	85	34	88
Cycle 19	66	85	47	94	29	86
Cycle 21	46	85	37	89	22	91
Cycle 23	31	84	20	100	16	94
Cycle 25	22	82	11	82	11	91

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; FACT-M=Functional Assessment of Cancer Therapy–Melanoma; VEM=vemurafenib.
*Includes instruments fully or partially completed.

RESULTS (continued)

Table 3. Patient Compliance on EORTC QLQ-C30 Through Cycle 25

Visit	COMBO450 (n=192)		ENCO300 (n=194)		VEM (n=191)	
	Patients Remaining in the Study (n)	Patients Who Filled Out the Instrument (%)*	Patients Remaining in the Study (n)	Patients Who Filled Out the Instrument (%)*	Patients Remaining in the Study (n)	Patients Who Filled Out the Instrument (%)*
Baseline	192	87	194	84	191	84
Cycle 3	186	96	186	92	182	91
Cycle 5	174	91	158	94	146	92
Cycle 7	161	91	132	94	110	94
Cycle 9	139	94	100	92	87	95
Cycle 11	121	92	86	88	63	86
Cycle 13	104	93	77	90	50	92
Cycle 15	94	89	72	86	40	95
Cycle 17	83	94	62	90	34	91
Cycle 19	66	86	47	87	29	93
Cycle 21	46	93	37	89	22	95
Cycle 23	31	90	20	95	16	94
Cycle 25	22	82	11	91	11	91

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; EORTC QLQ-30=European Organization for Research and Treatment of Cancer's core Quality of Life Questionnaire; VEM=vemurafenib.
*Includes instruments fully or partially completed.

FACT-M Melanoma Subscale

- Mean (SEM) changes from baseline scores for the FACT-M melanoma subscale over time are depicted in **Figure 2**.
 - At cycle 7 (after approximately 6 months of treatment), mean (SEM) changes from baseline were 1.35 (0.88), −2.92 (0.68), and −2.19 (0.94) in the COMBO450, ENCO300, and VEM arms, respectively.
 - At cycle 15 (after approximately 1 year of treatment), mean (SEM) changes from baseline were 0.40 (0.96), −2.33 (0.94), and −1.72 (1.8) in the COMBO450, ENCO300, and VEM arms, respectively.

Figure 2. Change From Baseline in FACT-M Melanoma Subscale Score Over Time

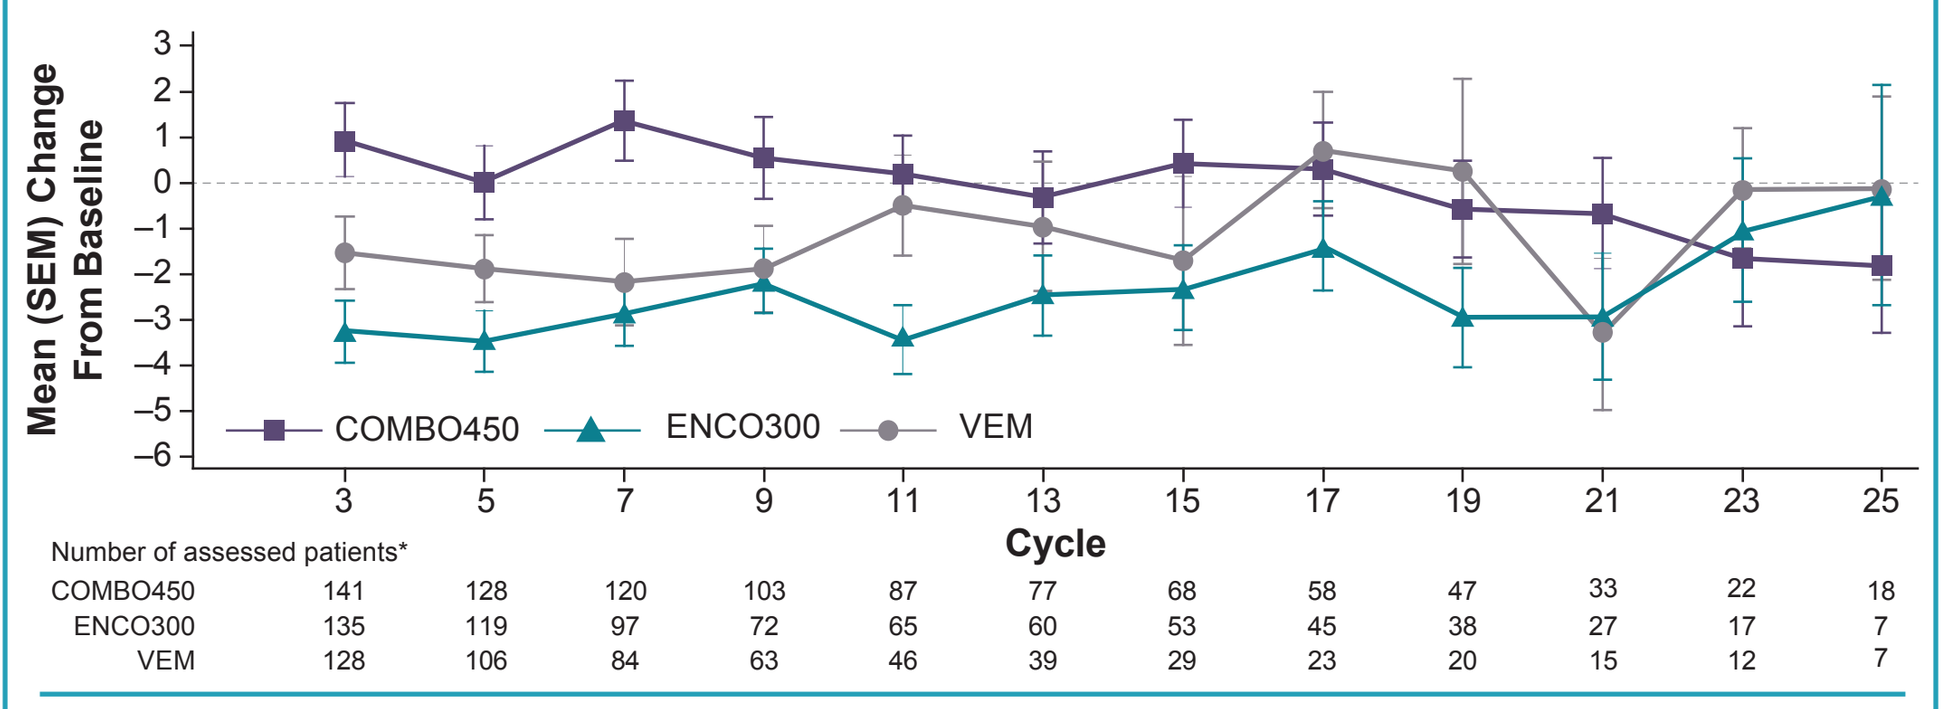
Mean (SEM) Change From Baseline
COMBO450
ENCO300
VEM
Cycle
Number of assessed patients*
COMBO450 141 128 120 103 87 77 68 58 47 33 22 18
ENCO300 135 119 97 72 65 60 53 45 38 27 17 7
VEM 128 106 84 63 46 39 29 23 20 15 12 7

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; FACT-M=Functional Assessment of Cancer Therapy–Melanoma; VEM=vemurafenib.
*Change from baseline determined in patients with non-missing baseline and post-baseline assessments.

- Patients in each arm were divided into tertiles by FACT-M melanoma subscale scores at baseline. Tertile 1 scores ranged from 22–51, tertile 2 scores ranged from 52–57, and tertile 3 scores ranged from 58–64.
 - Changes in FACT-M melanoma subscale score by tertile for each arm are depicted in **Figure 3**, which suggests that the highest improvement/maintenance of HRQoL was observed in the COMBO450 arm.
 - Improvement in HRQoL appears to be highest in patients with the lowest FACT-M melanoma subscale scores at baseline.

Figure 3. Change in FACT-M Melanoma Subscale Scores at Cycle 7 and Cycle 15 by Baseline FACT-M Score Tertiles in Each Treatment Arm

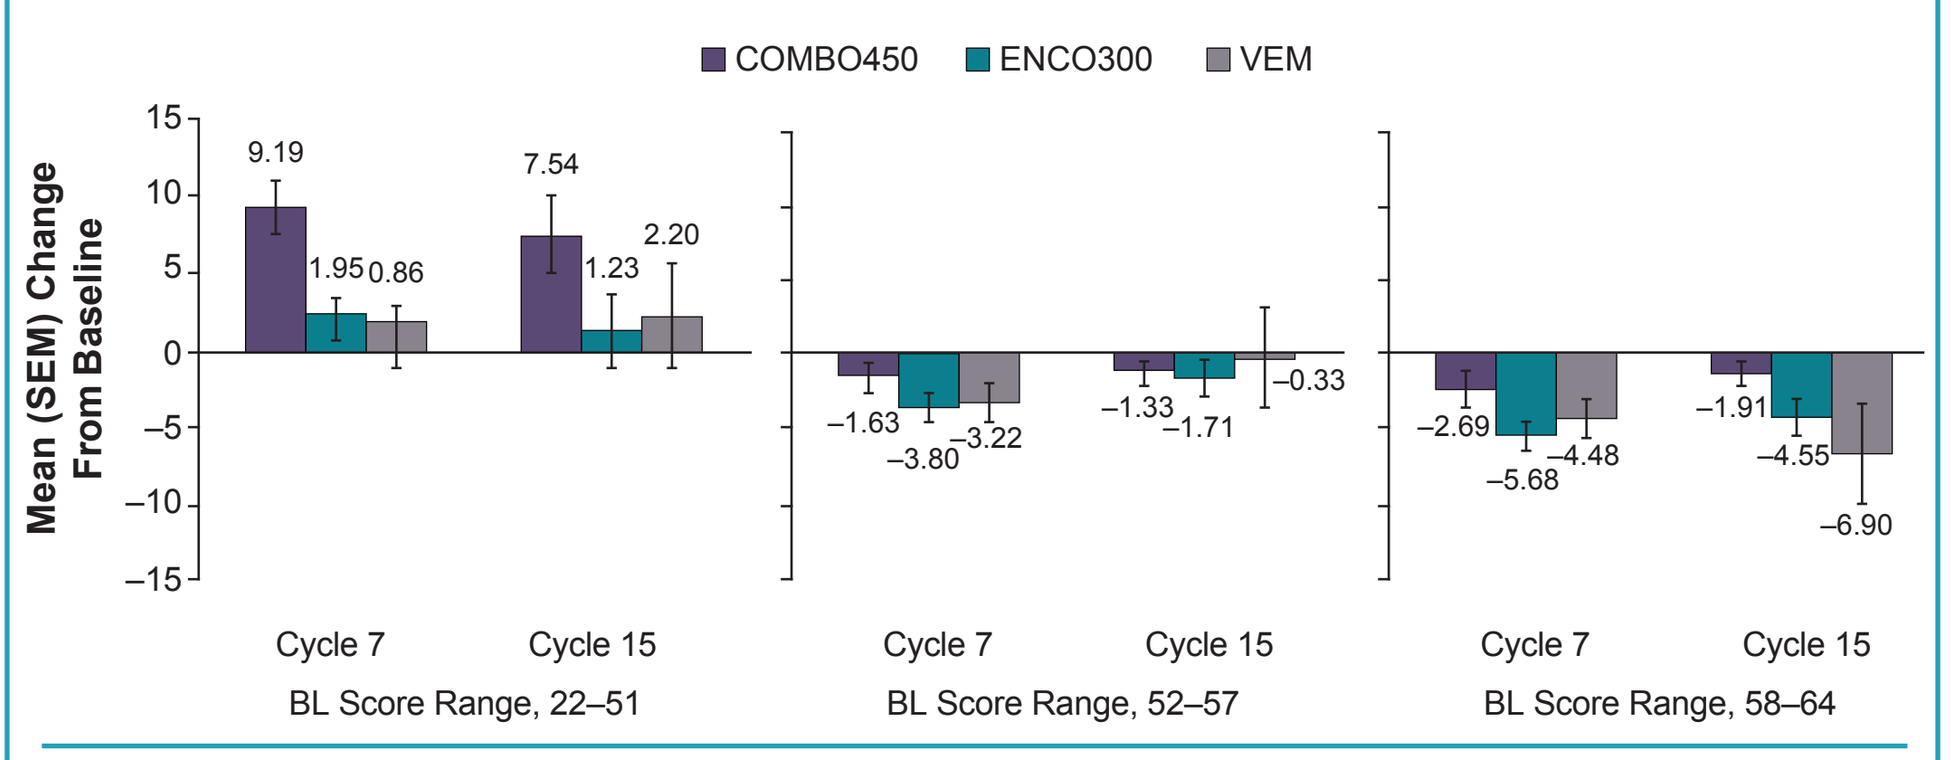
COMBO450
ENCO300
VEM
Mean (SEM) Change From Baseline
9.19
1.95
0.86
7.54
1.23
2.20
−1.63
−3.80
−3.22
−1.33
−1.71
−0.33
−2.69
−5.68
−4.48
−1.91
−4.55
−6.90
Cycle 7
Cycle 15
BL Score Range, 22–51
BL Score Range, 52–57
BL Score Range, 58–64

BL=baseline; COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; FACT-M=Functional Assessment of Cancer Therapy–Melanoma; VEM=vemurafenib.

- The median time to definitive 10% deterioration on the FACT-M melanoma subscale was not reached in the COMBO450 arm; it was 20.3 months (95% CI, 15.0, NE) in ENCO300 arm and 22.1 months (95% CI, 15.2, NE) in the VEM arm **(Figure 4)**.

Figure 4. Kaplan-Meier Plot of Time to Definitive 10% Deterioration in FACT-M Melanoma Subscale

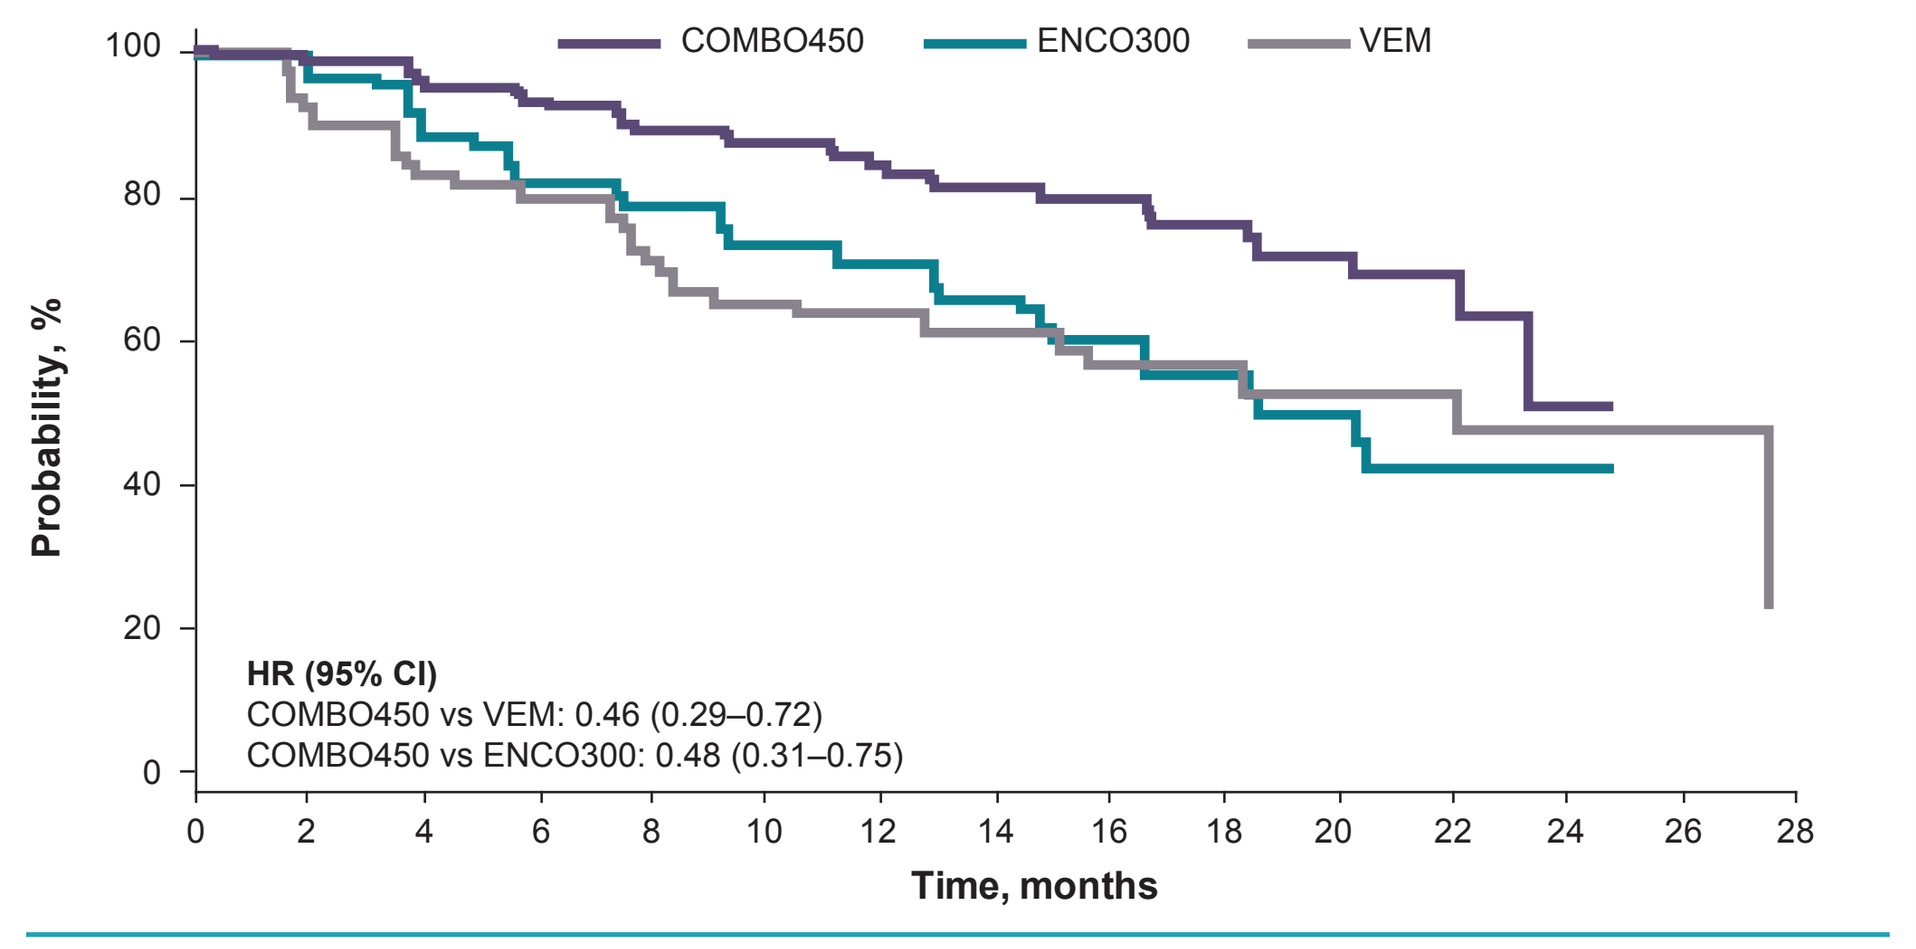
COMBO450
ENCO300
VEM
Probability, %
HR (95% CI)
COMBO450 vs VEM: 0.46 (0.29–0.72)
COMBO450 vs ENCO300: 0.48 (0.31–0.75)
Time, months

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; FACT-M=Functional Assessment of Cancer Therapy–Melanoma; HR=hazard ratio; VEM=vemurafenib.

EORTC QLQ-C30 Global Health Status

- Mean (SEM) change from baseline scores for the EORTC QLQ-C30 global health status scale over time are depicted in **Figure 5**.
 - At cycle 7 (after approximately 6 months of treatment), mean (SEM) changes from baseline were 1.53 (2.08), −8.74 (2.37), and −3.04 (3.06) in the COMBO450, ENCO300, and VEM arms, respectively.
 - At cycle 15 (after approximately 1 year of treatment), mean (SEM) changes were 5.02 (2.32), −10.03 (3.04), and −10.3 (5.54) in the COMBO450, ENCO300, and VEM arms, respectively.

Figure 5. Change From Baseline in EORTC QLQ-C30 Global Health Status Score Over Time

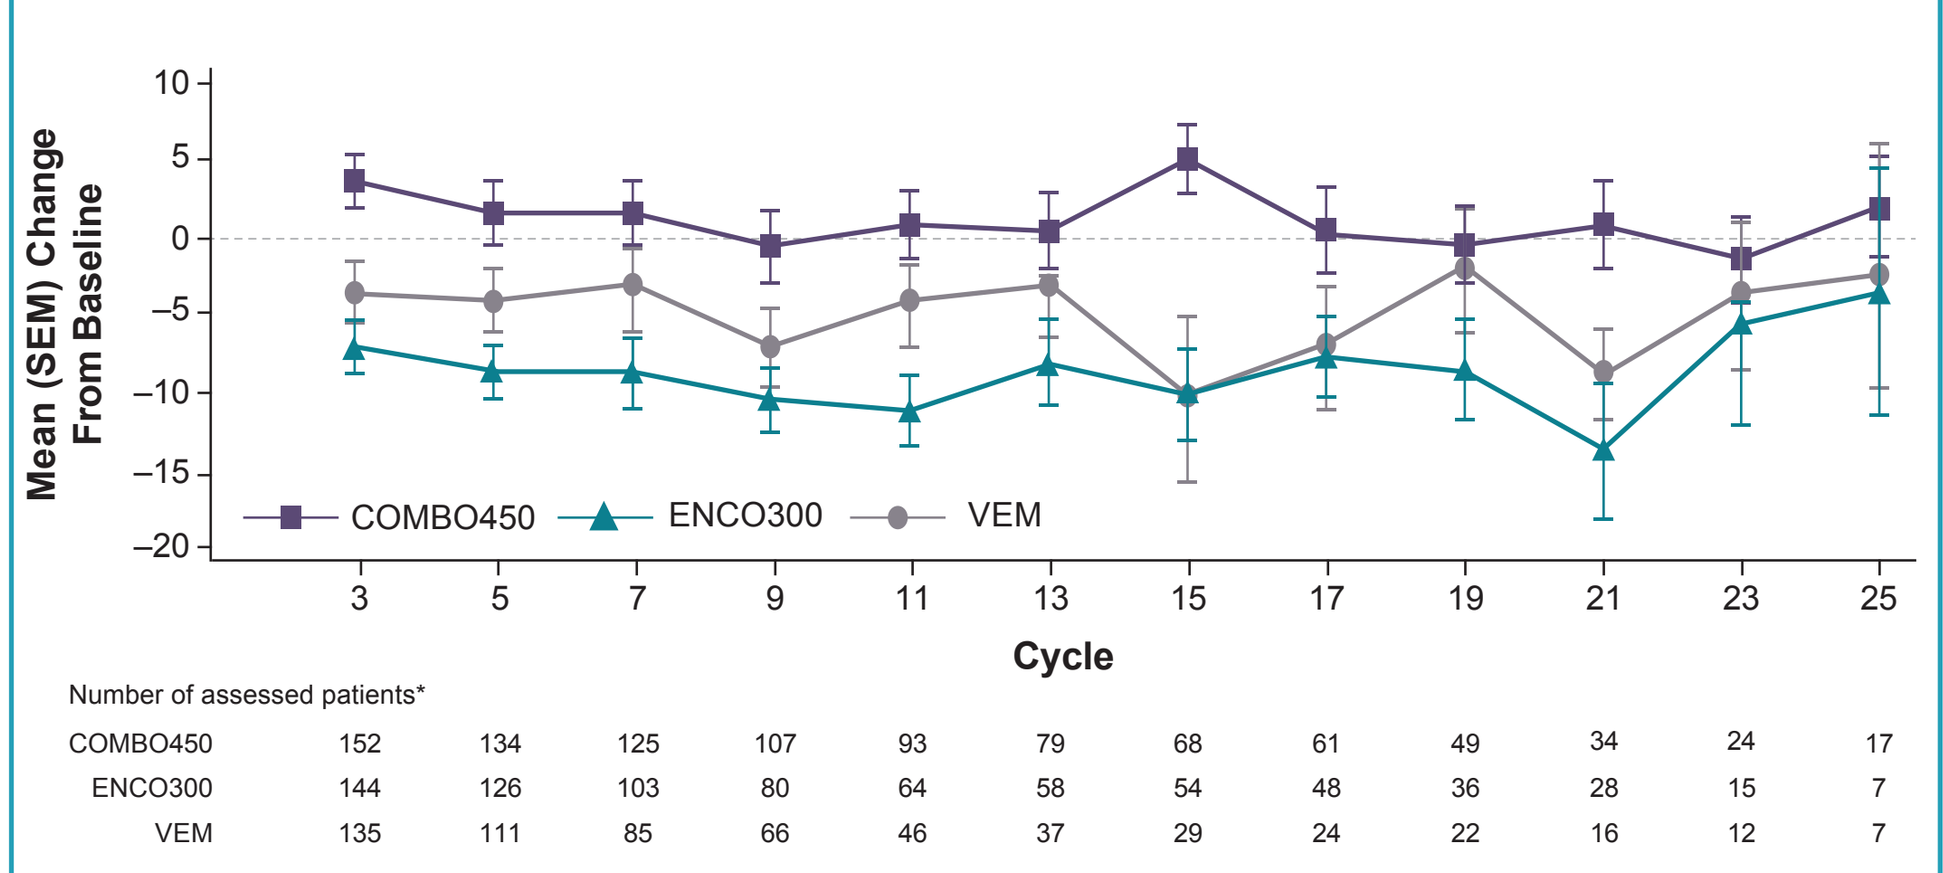
Mean (SEM) Change From Baseline
COMBO450
ENCO300
VEM
Cycle
Number of assessed patients*
COMBO450 152 134 125 107 93 79 68 61 49 34 24 17
ENCO300 144 126 103 80 64 58 54 48 36 28 15 7
VEM 135 111 85 66 46 37 29 24 22 16 12 7

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; EORTC QLQ-C30=European Organisation for Research and Treatment of Cancer core quality-of-life questionnaire; VEM=vemurafenib
*Change from baseline determined in patients with non-missing baseline and post-baseline assessments.

- Patients in each arm were divided into tertiles by EORTC QLQ-C30 global health status scores at baseline. Tertile 1 scores ranged from 0–50, tertile 2 scores ranged from 51–75, and tertile 3 scores ranged from 76–100.
 - Scores in the highest tertile generally correspond to normative scores from a general healthy adult population.[8]
 - Changes in EORTC QLQ-C30 global health status score by tertile for each arm are depicted in **Figure 6**, which suggests that the highest improvement/maintenance of HRQoL was observed in the COMBO450 arm.
 - Improvement in HRQoL appears to be highest in patients with the lowest EORTC QLQ-C30 global health status scores at baseline.
- Median times to definitive 10% deterioration in the global health status scale score were 23.9 months (95% CI, 20.4–NE), 14.7 months (95% CI, 0.2–18.4), and 16.6 months (95% CI, 11.9–NE) in the COMBO450, ENCO300, and VEM arms, respectively **(Figure 7)**.

Figure 6. Change in EORTC QLQ-C30 Global Health Status Score at Cycle 7 and Cycle 15 by Baseline EORTC QLQ-C30 Global Health Status Score Tertiles in Each Treatment Arm

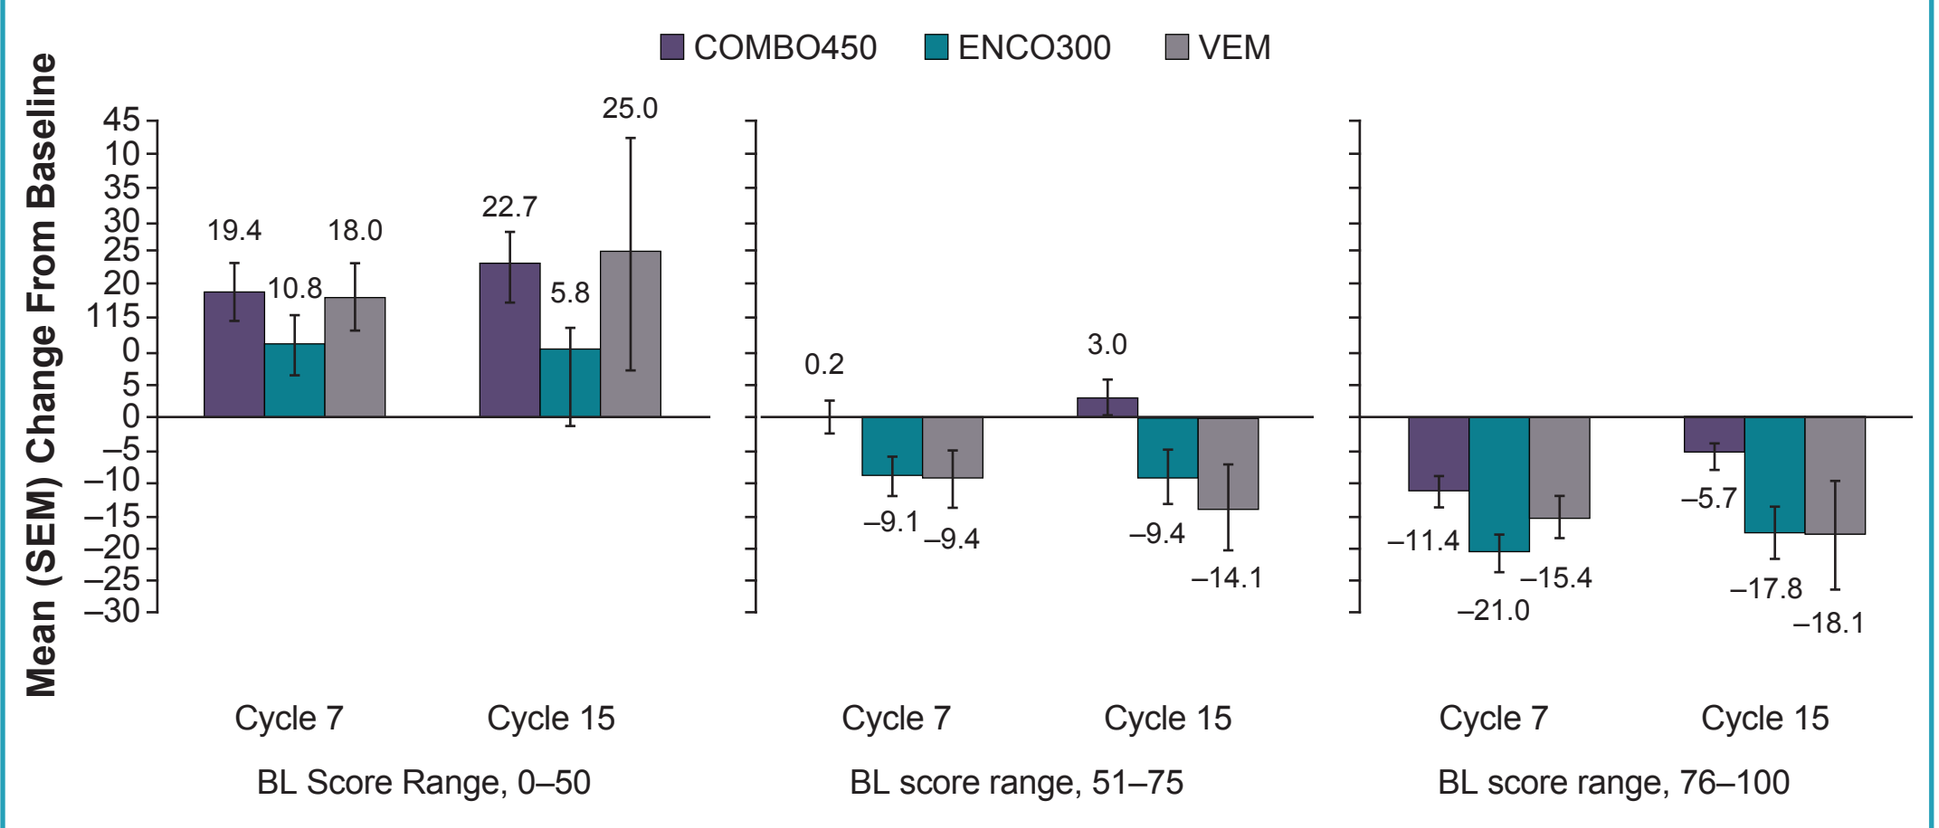
COMBO450
ENCO300
VEM
Mean (SEM) Change From Baseline
19.4
10.8
18.0
22.7
5.8
25.0
0.2
−9.1
−9.4
3.0
−9.4
−14.1
−11.4
−21.0
−15.4
−5.7
−17.8
−18.1
Cycle 7
Cycle 15
BL Score Range, 0–50
BL score range, 51–75
BL score range, 76–100

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; EORTC QLQ-C30=European Organisation for Research and Treatment of Cancer core quality-of-life questionnaire; VEM=vemurafenib.

Figure 7. Kaplan-Meier Plot of Time to Definitive 10% Deterioration in EORTC QLQ-C30 Global Health Status Scale

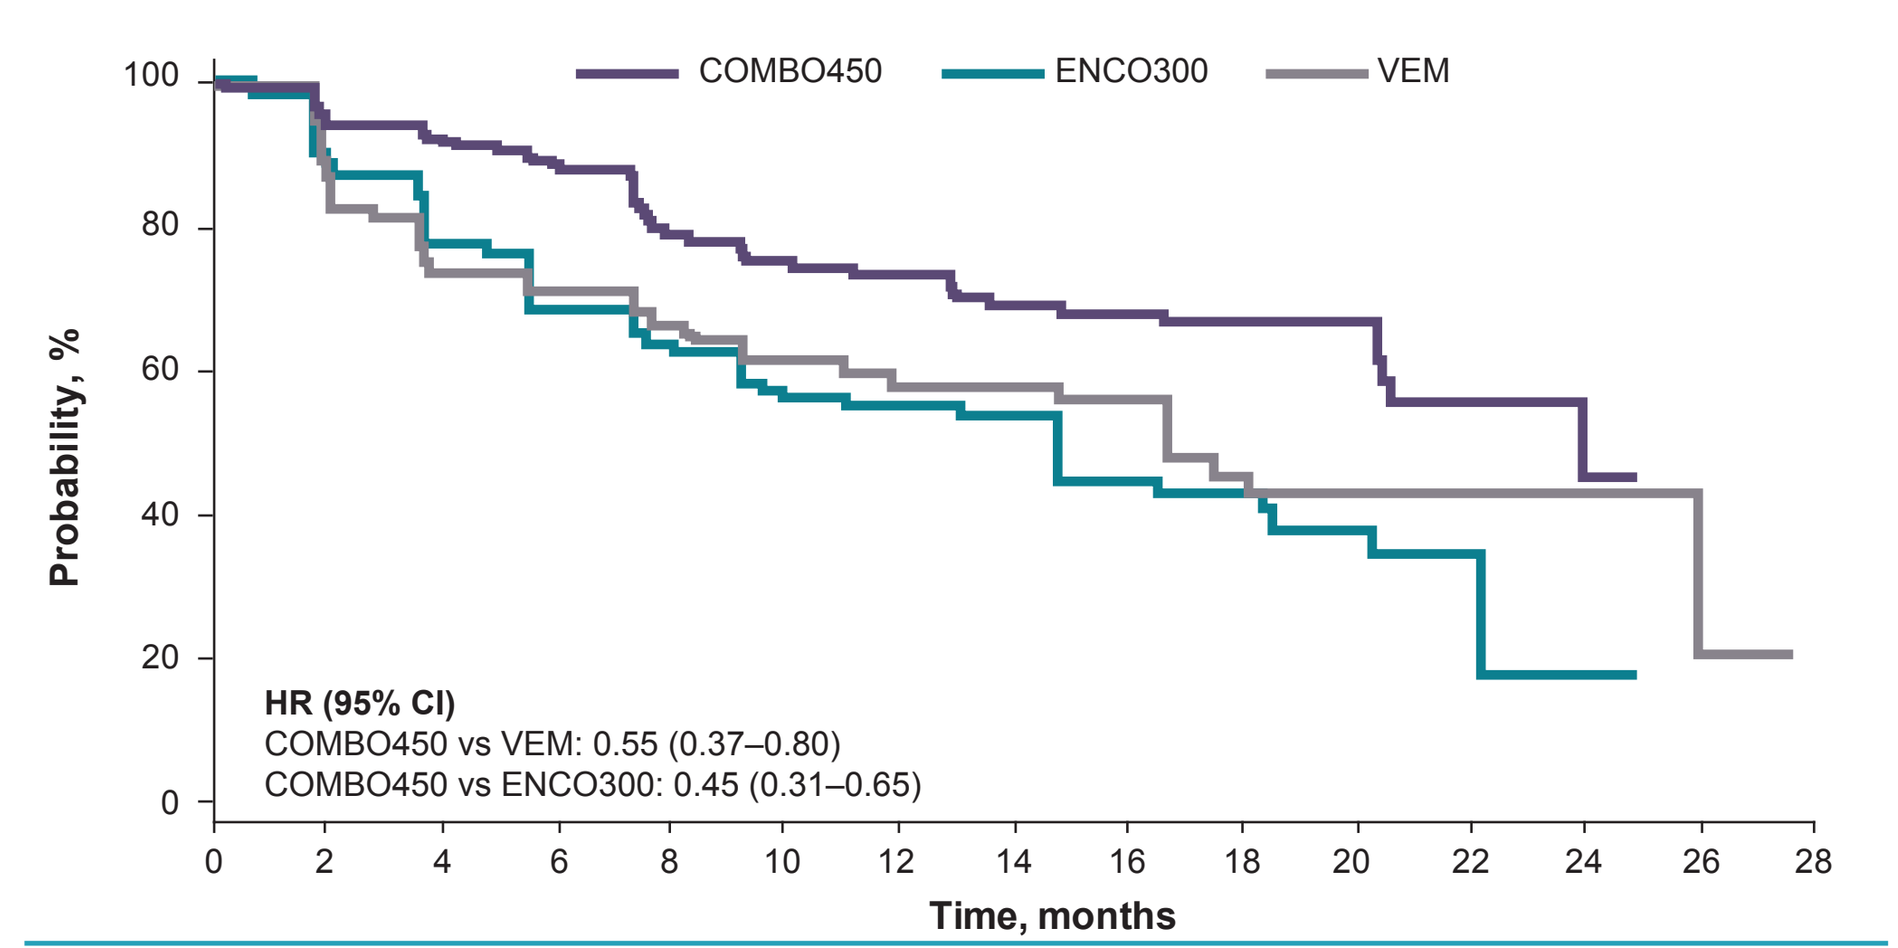
COMBO450
ENCO300
VEM
Probability, %
HR (95% CI)
COMBO450 vs VEM: 0.55 (0.37–0.80)
COMBO450 vs ENCO300: 0.45 (0.31–0.65)
Time, months

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; EORTC QLQ-C30=European Organisation for Research and Treatment of Cancer core quality-of-life questionnaire; HR=hazard ratio; VEM=vemurafenib.

HRQoL Change in Patients by Normal or High LDH Levels

- Mean (SEM) FACT-M melanoma subscale scores at baseline were similar in patients with normal baseline LDH levels (53.4 [0.4]; n=353) and high baseline LDH levels (49.4 [0.9]; n=128).
- Mean (SEM) EORTC QLQ-C30 global health status scores at baseline were higher (indicating better HRQoL) in patients with normal baseline LDH levels (68.9 [1.1]; n=358) than with high baseline LDH levels (57.4 [2.0]; n=130).
- Patients with high baseline LDH levels showed higher improvements on the FACT-M melanoma subscale and EORTC QLQ-C30 global health status scale at cycle 7 and cycle 15 than did those with normal baseline LDH levels **(Figure 8)**.
 - COMBO450 was associated with more improvement and maintenance of HRQoL than were the other treatment arms.

Figure 8. Change From Baseline at Cycles 7 and 15 by Baseline LDH Level of Normal or High

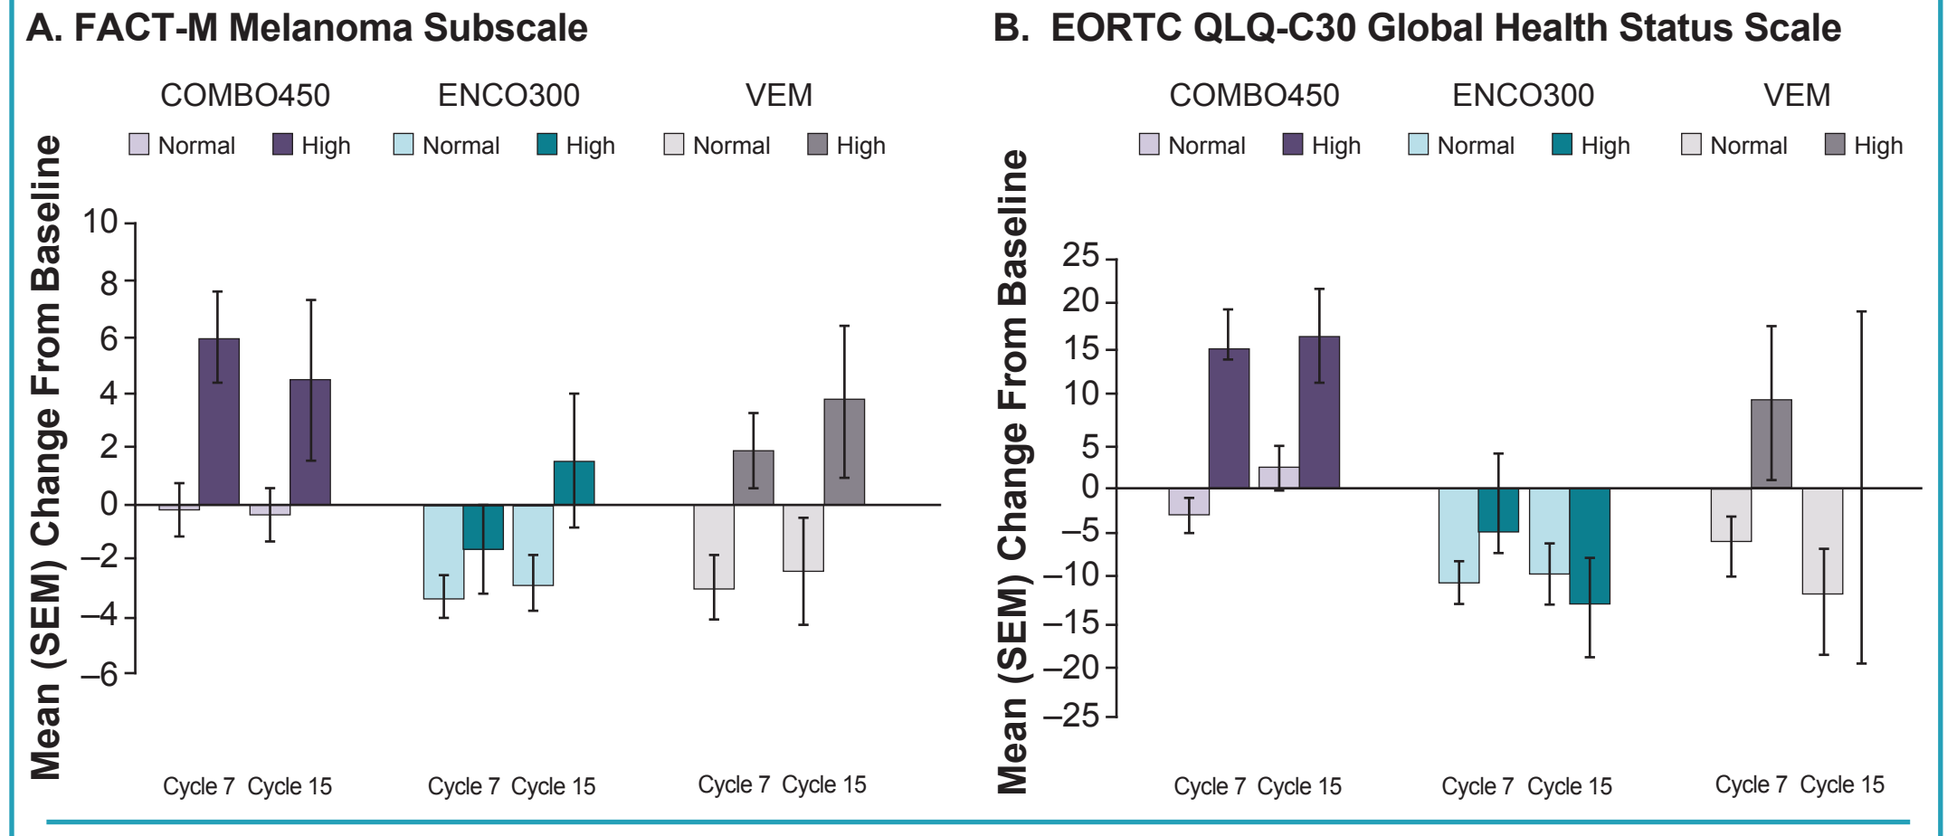
A. FACT-M Melanoma Subscale
B. EORTC QLQ-C30 Global Health Status Scale
COMBO450
ENCO300
VEM
Normal
High
Mean (SEM) Change From Baseline
Cycle 7
Cycle 15

COMBO450=encorafenib 450 mg once daily + binimetinib 45 mg twice daily; ENCO300=encorafenib 300 mg once daily; EORTC QLQ-C30=European Organisation for Research and Treatment of Cancer core quality-of-life questionnaire; FACT-M=Functional Assessment of Cancer Therapy–Melanoma; QLQ-C30=European Organisation for Research and Treatment of Cancer core quality-of-life questionnaire; LDH= lactate dehydrogenase; VEM=vemurafenib.

CONCLUSIONS

- Patient-reported HRQoL was rated consistently better with COMBO450 vs VEM or ENCO300 monotherapy.
 - After approximately 6 and 12 months of treatment, scores on both the FACT-M melanoma subscale and EORTC QLQ-C30 global health status scale were higher in the COMBO450 arm than in the ENCO300 or VEM arms.
- HRQoL was maintained with COMBO450 compared with ENCO300 or VEM, reflected in relative median times to definitive 10% deterioration on the FACT-M melanoma subscale and EORTC QLQ-C30 global health status.
- As reflected by changes in FACT-M melanoma subscale and EORTC scores over time, HRQoL was rated highest by patients who had the lowest FACT-M melanoma subscale and EORTC global health status scores at baseline or had worse prognosis (ie, high baseline LDH levels). The highest ratings were associated with treatment with COMBO450.

REFERENCES

1. Dummer R, et al. *Ann Oncol.* 2015;26(suppl 5):v126-132.
2. Schadendorf D, et al. *Eur J Cancer.* 2015;51(7):833-840.
3. Grob JJ, et al. *Lancet Oncol.* 2015;16(13):1389-1398.
4. Ascierto PA, et al. *Lancet Oncol.* 2016;17(9):1248-1260.
5. Dummer R, et al. Results of COLUMBUS Part 1: a phase 3 trial of encorafenib (ENCO) plus binimetinib (BINI) versus vemurafenib (VEM) or ENCO in BRAF-mutant melanoma. Presented at: Society for melanoma Research; November 6–9, 2016; Boston, MA.
6. Aaronson NK, et al. *J Natl Cancer Inst.* 1993;85(5):365-376.
7. Bedikian AY, et al. *Cancer Invest.* 2008;26(6):624-633.
8. Hinz A, et al. *Acta Oncol.* 2014;53(7):958-965.

DISCLOSURES

Helen J. Gogas: Consultant for Roche, BMS, MSD, Novartis, and Amgen

Reinhard Dummer: Honoraria from and consulting/advisory role for Roche, BMS, GSK, MSD, Novartis, and Amgen; research funding from Roche, BMS, GSK, MSD, and Novartis

Paolo A. Ascierto: Consulting fees from BMS, Roche/Genentech, MSD, Novartis, Amgen, Array BioPharma, Merck Serono, Pierre Fabre, and Incyte; research funding from BMS, Roche/Genentech, and Array BioPharma.

Ana Arance: Honoraria from and consulting/advisory role and speakers bureau for Novartis, Roche, MSD, and BMS; travel expenses from Roche and BMS

Mario Mandala: Honoraria from Novartis, GSK, BMS, MSD, and Roche; speakers bureau for Novartis, GSK, Roche, and BMS; advisory board member for Novartis, Amgen, MSD, and BMS; research funding from Roche

Gabriella Liszkay: Nothing to disclose

Claus Garbe: Honoraria and travel expenses from and served in a consulting/advisory role and speakers bureau member for Amgen, BMS, MSD, Novartis, Roche, and Philogen; has received research funding for University Hospital Tübingen from BMS, Novartis, and Roche

Dirk Schadendorf: Honoraria and travel expenses from and consulting/advisory role and speakers bureau for Amgen, BMS, Novartis, Roche, and MSD; research funding for University Hospital Essen from Amgen, BMS, Novartis, Roche, and MSD

Ivana Krajsová: Advisory board member for BMS, Novartis, Roche, MSD; travel expenses from BMS and MSD

Ralf Gutzmer: Consulting fees and/or honoraria from Roche, BMS, MSD, GSK, Novartis, Almirall, LEO, Amgen, Pfizer, Pierre Fabre; research funding from Roche, Novartis, Pfizer, Johnson & Johnson; travel expenses from BMS, Roche.

Vanna Chiarion Sileni: Honoraria received from Novartis, GSK, BMS, MSD, and Roche; speakers bureau for Novartis, GSK, Roche, and BMS; advisory board member for Novartis, Amgen, MSD, BMS, and Roche

Caroline Dutriaux: Nothing to disclose

Jan Willem B. de Groot: Consulting/advisory role for Amgen, Bayer, Celgene, Roche, BMS, GSK, MSD, and Merck Serono

Naoya Yamazaki: Advisory role for Chugai Pharma, Bristol-Myers Squibb Japan, and Ono Pharmaceutical; honoraria from Chugai Pharma, Bristol-Myers Squibb Japan, Ono Pharmaceutical, GSK, Takeda, AstraZeneca Japan, Boehringer Ingelheim, and Maruh

Carmen Loquai: Advisory board member for Roche, Novartis, BMS, MSD, Biontech, Amgen, and Pierre Fabre; speakers fees from Roche, Novartis, BMS, and MSD; travel expenses from Roche, Novartis, BMS, MSD, and Amgen

Laure A. de Parseval: Employee of Novartis Pharma AG; may own stock and/or stock options

Michael Pickard: Employee of Array BioPharma; may own stock and/or stock options

Victor Sandor: Employee/leadership role at Array BioPharma; stock and/or other ownership of Array BioPharma and Incyte Corp

Caroline Robert: Consultant for Roche, Novartis, BMS, MSD, and Amgen

Keith Flaherty: Honoraria from and consulting/advisory role for Novartis and Array BioPharma; research funding from Novartis

Available at:
www.arraybiopharma.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Acknowledgments

Editorial support for development of this manuscript was provided by Bret Fulton at Complete Healthcare Communications, LLC (West Chester, PA), a CHC Group company, and funded by Array BioPharma Inc.

This study is sponsored by Array BioPharma Inc.