688

Gastrointestinal Cancers Symposium (ASCO GI)
January 17–19, 2019
San Francisco, CA

Updated Results of the BEACON Colorectal Cancer (CRC) Safety Lead-In: Encorafenib (ENCO) + Binimetinib (BINI) + Cetuximab (CETUX) for *BRAF* V600E–Mutant CRC

Scott Kopetz,[1] Axel Grothey,[2] Rona Yaeger,[3] Pieter-Jan Cuyle,[4] Sanne Huijberts,[5] Jan H. M. Schellens,[5] Elena Elez,[6] Marwan Fakih,[7] Clara Montagut,[8] Marc Peeters,[9] Jayesh Desai,[10] Takayuki Yoshino,[11] Fortunato Ciardiello,[12] Harpreet Wasan,[13] Victor Sandor,[14] Kati Maharry,[14] Janna Christy-Bittel,[14] Ashwin Gollerkeri,[14] Eric Van Cutsem,[15] Josep Tabernero[16]

[1]UT MD Anderson Cancer Center, Houston, TX, USA; [2]West Cancer Center, University of Tennessee, Germantown, TN, USA; [3]Memorial Sloan-Kettering Cancer Center, New York, NY, USA; [4]Imelda Ziekenhuis, Bonheiden, Belgium; [5]The Netherlands Cancer Institute, Amsterdam, the Netherlands; [6]Vall d'Hebron Institute of Oncology, Barcelona, Spain; [7]City of Hope, Duarte, CA, USA; [8]Department of Medical Oncology, Hospital Universitari del Mar, Barcelona, Spain; [9]Antwerp University Hospital, Edegem, Belgium; [10]Royal Melbourne Hospital and Peter MacCallum Cancer Centre, Walter and Eliza Hall Institute, Parkville, VIC, Australia; [11]National Cancer Center Hospital East, Kashiwa, Japan; [12]University of Campania, Naples, Italy; [13]Hammersmith Hospital, Department of Cancer Medicine, London, UK; [14]Array BioPharma Inc, Boulder, CO, USA; [15]University Hospitals Gasthuisberg Leuven and KU Leuven, Leuven, Belgium; [16]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology, Barcelona, Spain*

BACKGROUND

- *BRAF* V600E mutation occurs in up to 15% of patients with metastatic colorectal cancer (mCRC), confers a poor prognosis, and results in worse outcomes with standard-of-care treatments for mCRC.[1-11]
- In a phase 2 study in *BRAF* V600E mCRC, treatment with the anti–epidermal growth factor receptor (EGFR) antibody cetuximab (CETUX) plus the *BRAF* inhibitor encorafenib (ENCO) led to a median progression-free survival (PFS) of 4.2 months, an overall response rate (ORR) of 24%, and a median overall survival (OS) of 9.3 months, which represent improvements in outcomes relative to historic data.[12]
- *BRAF* inhibitors may cause feedback activation of EGFR in *BRAF*-mutant colorectal cancer, leading to continued cell proliferation.[12-14] It is hypothesized that this feedback activation may be overcome by targeting additional points in the pathway, such as MEK.[15,16]
- BEACON CRC (NCT02928224) is a randomized, open-label, 3-arm Phase 3 study evaluating binimetinib (BINI), a MEK inhibitor, plus ENCO and CETUX compared with chemotherapy plus CETUX and ENCO plus CETUX in patients with *BRAF* V600E mCRC after 1 or 2 prior metastatic treatment regimens (Figure 1).[17]
- As part of the BEACON CRC trial, a safety lead-in (SLI) phase was conducted in 30 patients to evaluate the safety and tolerability of the triplet regimen and to assess preliminary efficacy prior to initiating the phase 3 portion of the study.
- Previously reported data showed promising safety in 30 patients treated and efficacy in 29 patients with *BRAF* V600E mCRC.[18]
- Here we report updated safety and efficacy results, including mature OS, from the SLI.

Figure 1. The BEACON Study Clinical Trial Design

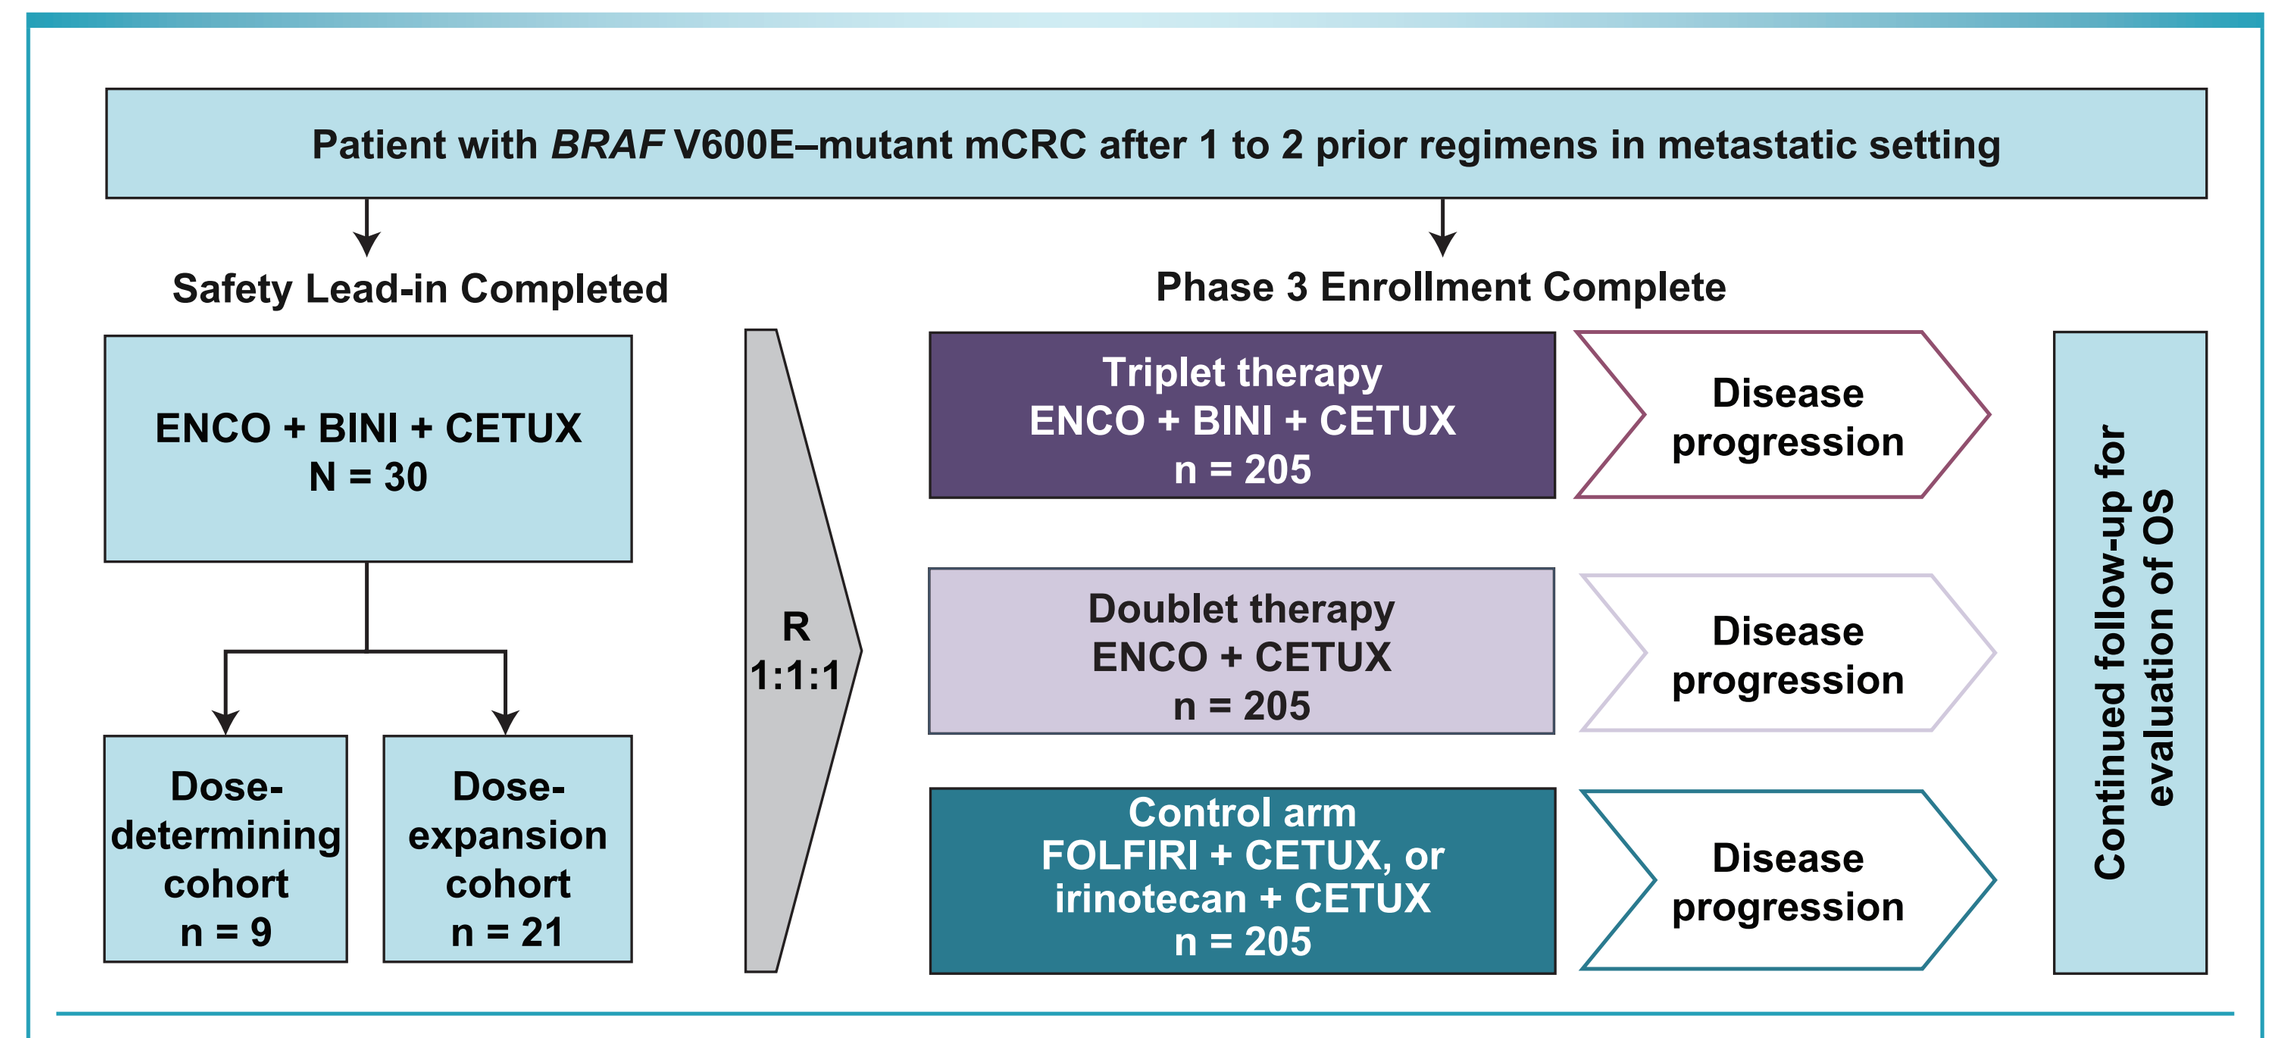

Patient with BRAF V600E–mutant mCRC after 1 to 2 prior regimens in metastatic setting
Safety Lead-in Completed
ENCO + BINI + CETUX
N = 30
Dose-determining cohort
n = 9
Dose-expansion cohort
n = 21
R
1:1:1
Phase 3 Enrollment Complete
Triplet therapy
ENCO + BINI + CETUX
n = 205
Doublet therapy
ENCO + CETUX
n = 205
Control arm
FOLFIRI + CETUX, or irinotecan + CETUX
n = 205
Disease progression
Disease progression
Disease progression
Continued follow-up for evaluation of OS

A separate Safety Lead-in cohort of n=7 in Japan was enrolled subsequently. Results will be reported at a later time.

OBJECTIVE

Study Population

- All patients who received at least one dose of study drug were included in the safety analyses (n=30).
- For efficacy analyses, all patients with a *BRAF* V600E mutation who received at least one dose of study drug were included.

Endpoints

- The primary endpoint of the SLI was the assessment of safety and tolerability, which included
 - Dose-limiting Toxicities (DLTs);
 - Incidence and severity of AEs and changes in clinical laboratory parameters, vital signs, ECGs, ECHO/MUGA scans, and ophthalmic examinations; and
 - Incidence of dose interruptions, dose modifications, and discontinuations.
- Efficacy endpoints included
 - Confirmed Overall Response Rate (ORR; per RECIST v1.1)
 - Duration of Response (DOR)
 - Progression Free Survival (PFS),
 - Time to Response (TTR), and
 - Overall Survival (OS)
- Radiographic assessment of tumor response and progression was determined locally by the investigator.
- Blinded central review of radiographically determined tumor response was also conducted retrospectively and reported.
- Pharmacokinetic endpoints were also evaluated and will be presented elsewhere.

METHODS

- All patients in the SLI received ENCO 300 mg once daily, BINI 45 mg twice daily, and CETUX at the standard weekly dose (400 mg/m^2, then 250 mg/m^2 once weekly)
- Safety data were reviewed by an independent data monitoring committee during the dose-determining cohort before proceeding to dose expansion and then following the completion of dose expansion (after enrollment of a total of 30 patients in the SLI).
- Efficacy was assessed on the basis of radiological imaging (eg, computed tomography, magnetic resonance imaging, x-ray, whole-body bone scans)
- Tumor response was determined by the investigator according to RECIST, version 1.1
 - Tumor assessment was performed every 6 weeks from the first dose for the first 24 weeks and then every 12 weeks until disease progression, withdrawal of consent for treatment, initiation of subsequent anticancer therapy, or lost to follow-up

Key Inclusion Criteria

- Patients ≥18 years of age with confirmed mCRC.
- Presence of a *BRAF* V600E mutation in tumor tissue as detected by central testing or by polymerase chain reaction (PCR) or next-generation sequencing (NGS) per local testing. Patients could enroll based on local determination of *BRAF* V600E mutation, however, confirmation by a central laboratory was required for all patients within 30 days of starting treatment.
- Progression after 1 to 2 prior regimens in the metastatic setting.
- Eligible to receive CETUX per local label with regard to tumor RAS status.
- Eastern Cooperative Oncology Group (ECOG) performance status (PS) of 0 or 1.

Key Exclusion Criteria

- Patients were excluded if they had previous treatment with any RAF or MEK inhibitor, cetuximab, panitumumab, or other EGFR inhibitor; symptomatic brain metastasis or leptomeningeal disease.

RESULTS

Patients

- From November 1, 2016, to April 24, 2017, a total of 30 patients were treated in the SLI portion of BEACON (Table 1).
 - The median age of the patients was 59 years (range, 38–77 years).
- 60% of patients had received 1 prior line of therapy, and 40% of patients received 2 prior lines of therapy.
 - 43% had received prior irinotecan.
- At the time of data cut-off (September 2, 2018), 6 patients remained on treatment.

Table 1. Baseline Demographics and Disease Characteristics

Characteristic	Patients (N=30)
BRAF V600E mutation, n (%)[a]	29 (97)
Male sex, n (%)	13 (43)
Race, n (%)	
White	29 (97)
Black or African American	1 (3)
Age, median (range), years	59 (38–77)
ECOG PS 0, n (%)	17 (57)
Location of primary tumor, n (%)	
Left side	9 (30)
Right side	18 (60)
Unknown	3 (10)
No. of organs with ≥2 / metastasis, n (%)	22 (73)
Metastatic site locations, n (%)	
Liver	20 (67)
Lymph nodes	15 (50)
Peritoneum	11 (37)
Lung	9 (30)
Other	15 (50)
Resection of primary tumor, n (%)	
Yes	21 (70)
No	9 (30)
No. of prior systemic therapies, n (%)[b]	
1	18 (60)
2	12 (40)
Received prior irinotecan, n (%)	13 (43)
MSI-H, n (%)[c]	1 (3)
CEA, median (range) at baseline, µg/mL	28 (1–3,434)

CEA, carcinoembryonic antigen; ECOG, Eastern Cooperative Oncology Group; MSI-H, microsatellite instability high. [a]1 patient had a non-V600E *BRAF* mutation. [b]Includes prior systemic therapies in the metastatic setting only. [c]Based on immunohistochemical assessment of MLH1 and MSH6.

Safety Profile

- All 30 patients received at least 1 drug treatment and were assessed for safety (Table 2).
- No unexpected toxicities occurred with this new triplet combination since the previous data analysis.
- DLTs were observed in 5 of 30 patients and have been reported previously.[18]
- A total of six (20%) patients had at least one drug discontinued due to AEs.
 - One (3%) discontinued all three drugs due to grade 2 fatigue;
 - Two (7%) discontinued binimetinib alone due to increased blood creatinine (n=1) and retinal detachment (n=1);
 - Two (7%) discontinued cetuximab alone due to an allergic reaction; and
 - One (3%) discontinued both encorafenib and binimetinib due to increased blood bilirubin.
 - The patient received a dose of cetuximab two weeks following discontinuation of encorafenib and binimetinib and then discontinued study treatment completely two weeks later due to clinical progression.
 - There were five (17%) on-treatment deaths, all due to disease progression.

RESULTS (continued)

Table 2. All-Grade (≥20%) or Grade 3/4 (≥10%) Adverse Events (N=30) Regardless of Causality

Event, n (%)	Any Grade	Grade 3 / 4
Diarrhea	23 (77)	1 (3)
Dermatitis acneiform	20 (67)	0
Fatigue	19 (63)	4 (13)
Nausea	19 (63)	2 (7)
Vomiting	15 (50)	2 (7)
Dry skin	15 (50)	0
Anemia	12 (40)	3 (10)
Decreased appetite	12 (40)	2 (7)
Abdominal pain	11 (37)	1 (3)
Increased creatinine kinase	11 (37)	3 (10)
Pyrexia	11 (37)	0
Dyspnea	10 (33)	2 (7)
Constipation	9 (30)	0
Arthralgia	8 (27)	0
Creatine increased	8 (27)	0
Skin fissure	8 (27)	0
Vision blurred	8 (27)	0
Increased aspartate aminotransferase	6 (20)	3 (10)
Asthenia	6 (20)	0
Malaise	6 (20)	1 (3)
Myalgia	6 (20)	0
PPED syndrome	6 (20)	0
Rash maculopapular	6 (20)	0
Urinary tract infection	5 (17)	3 (10)

Efficacy

- Efficacy was evaluated in n=29 patients with *BRAF* V600E mutations
 - A non-V600 *BRAF* mutation (G466V) was identified in 1 patient, who was excluded from the efficacy analysis.
 - Median time of 7.9 months (range, 1.0–21.4 months) on study treatment (Table 3).
- Confirmed ORR by local assessment was 48% (14/29 patients), including complete responses (CRs) in 3 patients (Figure 2A). The duration of response per Kaplan-Meier estimate was ≥6 months in 43% of responders.
 - ORR in patients with 1 previous line of therapy, 59% (10/17; 8 partial responses [PRs] and 2 CRs)
 - ORR in patients with 2 previous lines of therapy, 33% (4/12; 3 PRs and 1 CR)
- Confirmed ORR by central assessment was 41% (12/29 patients) with 2 patients reported as CR; Figure 2B). The duration of response was ≥6 months in 73% of responders.
 - ORR in patients with 1 previous line of therapy, 53% (9/17; 8 partial responses [PRs] and 1 CRs)
 - ORR in patients with 2 previous lines of therapy, 25% (3/12; 2 PRs and 1 CR)

Figure 2A. Best Percentage Change in Tumor Measurements From Baseline in Patients With *BRAF* V600E mCRC – Local Assessment

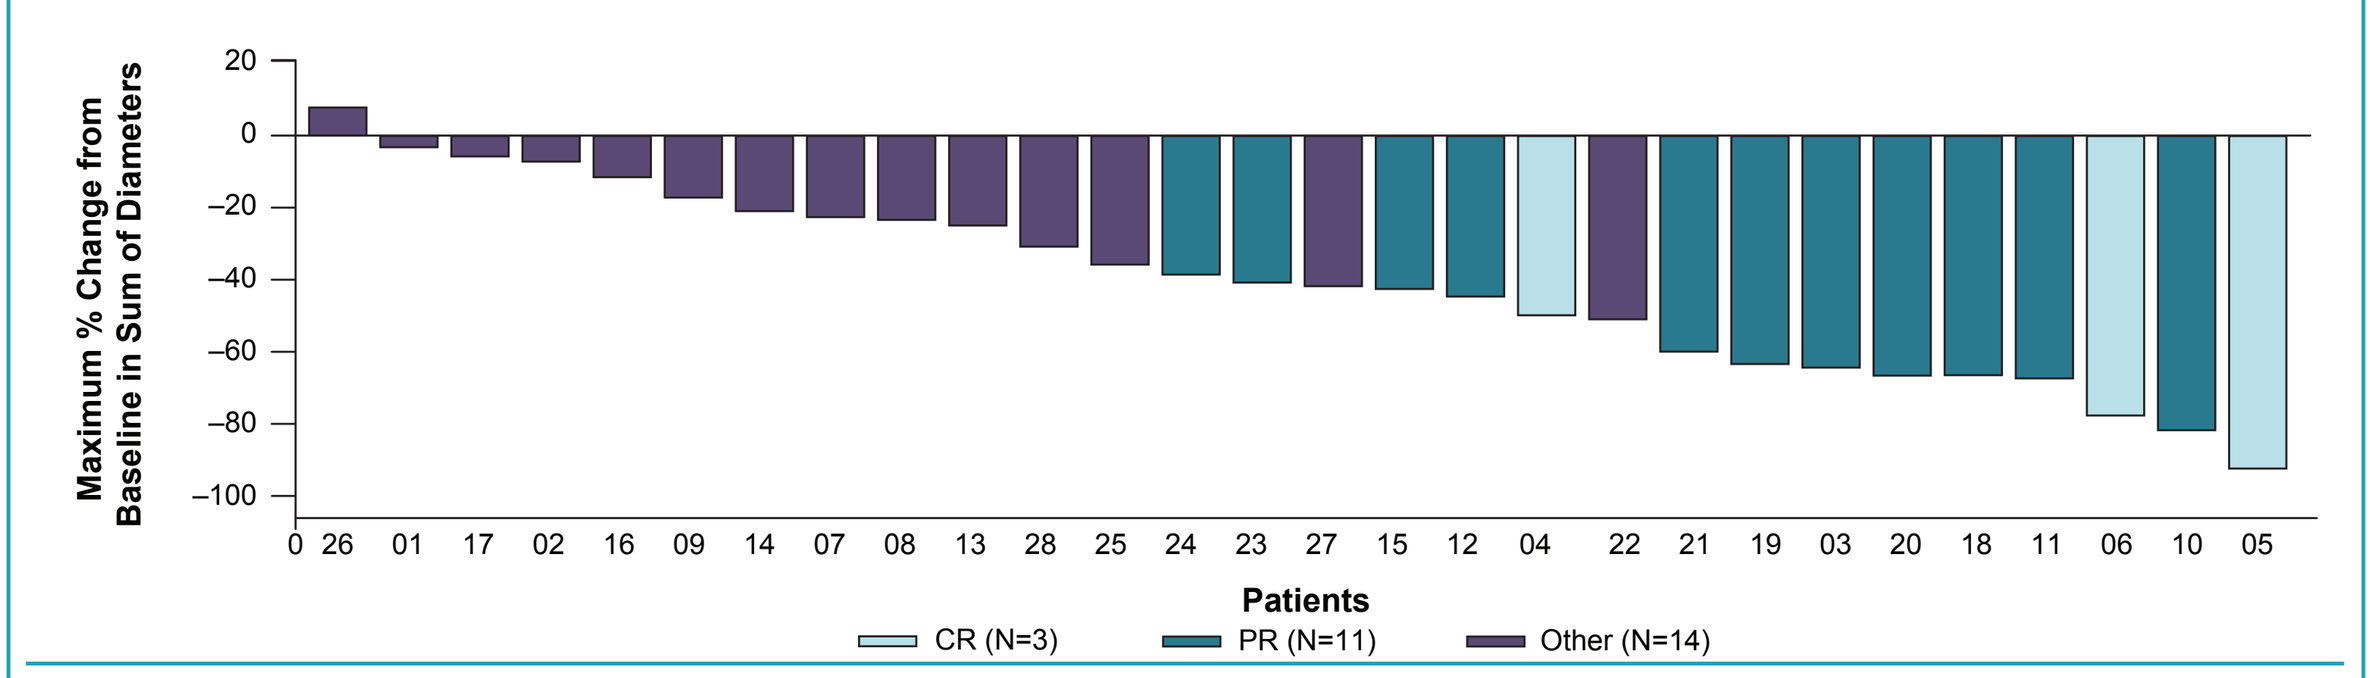

Maximum % Change from Baseline in Sum of Diameters
20
0
−20
−40
−60
−80
−100
0 26 01 17 02 16 09 14 07 08 13 28 25 24 23 27 15 12 04 22 21 19 03 20 18 11 06 10 05
Patients
CR (N=3)
PR (N=11)
Other (N=14)

One patient was without a postbaseline sum of diameters (not presented). Colors represent best response (confirmed). Patients with CR, defined as the disappearance of all target lesions, can have pathological lymph node metastases present; target or nontarget lymph node metastases must have reduction in short axis to < 10 mm. One patient had SD that was too early for assessment and was classified as NE. NE, not evaluable.

Figure 2B. Best Percentage Change in Tumor Measurements From Baseline in Patients With *BRAF* V600E mCRC – Central Assessment

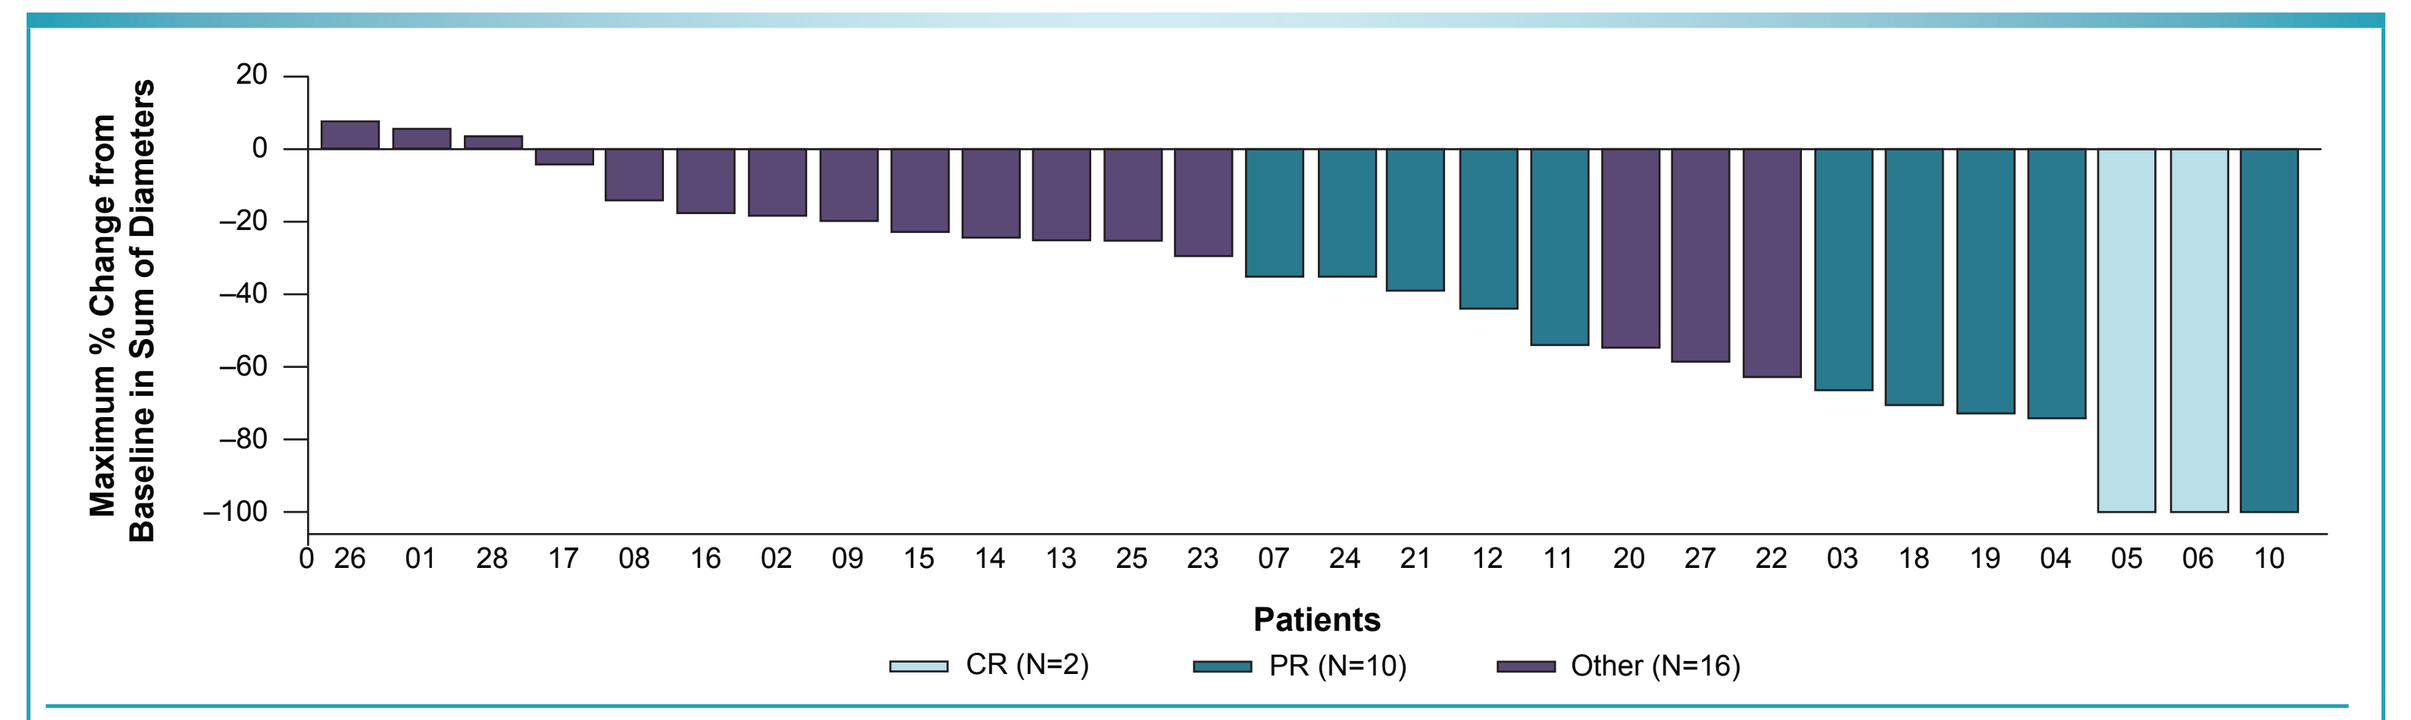

Maximum % Change from Baseline in Sum of Diameters
20
0
−20
−40
−60
−80
−100
0 26 01 28 17 08 16 02 09 15 14 13 25 23 07 24 21 12 11 20 27 22 03 18 19 04 05 06 10
Patients
CR (N=2)
PR (N=10)
Other (N=16)

One patient was without a postbaseline sum of diameters (not presented). Colors represent best response (confirmed). Patients with CR, defined as the disappearance of all target lesions, can have pathological lymph node metastases present; target or nontarget lymph node metastases must have reduction in short axis to < 10 mm. One patient had SD that was too early for assessment and was classified as NE. NE, not evaluable.

Table 3. Summary of Confirmed Best Overall Response

Confirmed Best Overall Response (BOR) n (%)[a]	Patients (N = 29)[b]	
	Local Assessment	Central Assessment
Overall Response Rate		
(CR + PR) [95% CI]	14 (48) [29–68%]	12 (41) [24–61%]
Complete Response (CR)	3 (10)	2 (7)
Partial Response (PR)	11 (38)	10 (34)
Stable Disease (SD)	13 (45)	13 (45)
Progressive Disease (PD)	0	0
Not Evaluable (NE)[c]	2 (7)	4 (14)

[a]Response confirmed per Response Evaluation Criteria in Solid Tumors, version 1.1. [b]Patients with *BRAF* V600E mutations only. [c]Nonresponders per intent-to-treat analysis. CR, Complete Response; PR, Partial Response; SD, Stable Disease; PD, Progressive Disease; NE, Not Evaluable

- Median duration of response was 5.5 months (95% CI, 4.1 months–not reached [NR]) per local assessment and 8.2 months (95% CI, 2.8 months–NR) per central assessment (Figure 3).

Figure 3. Duration on Treatment in Patients With *BRAF* V600E mCRC

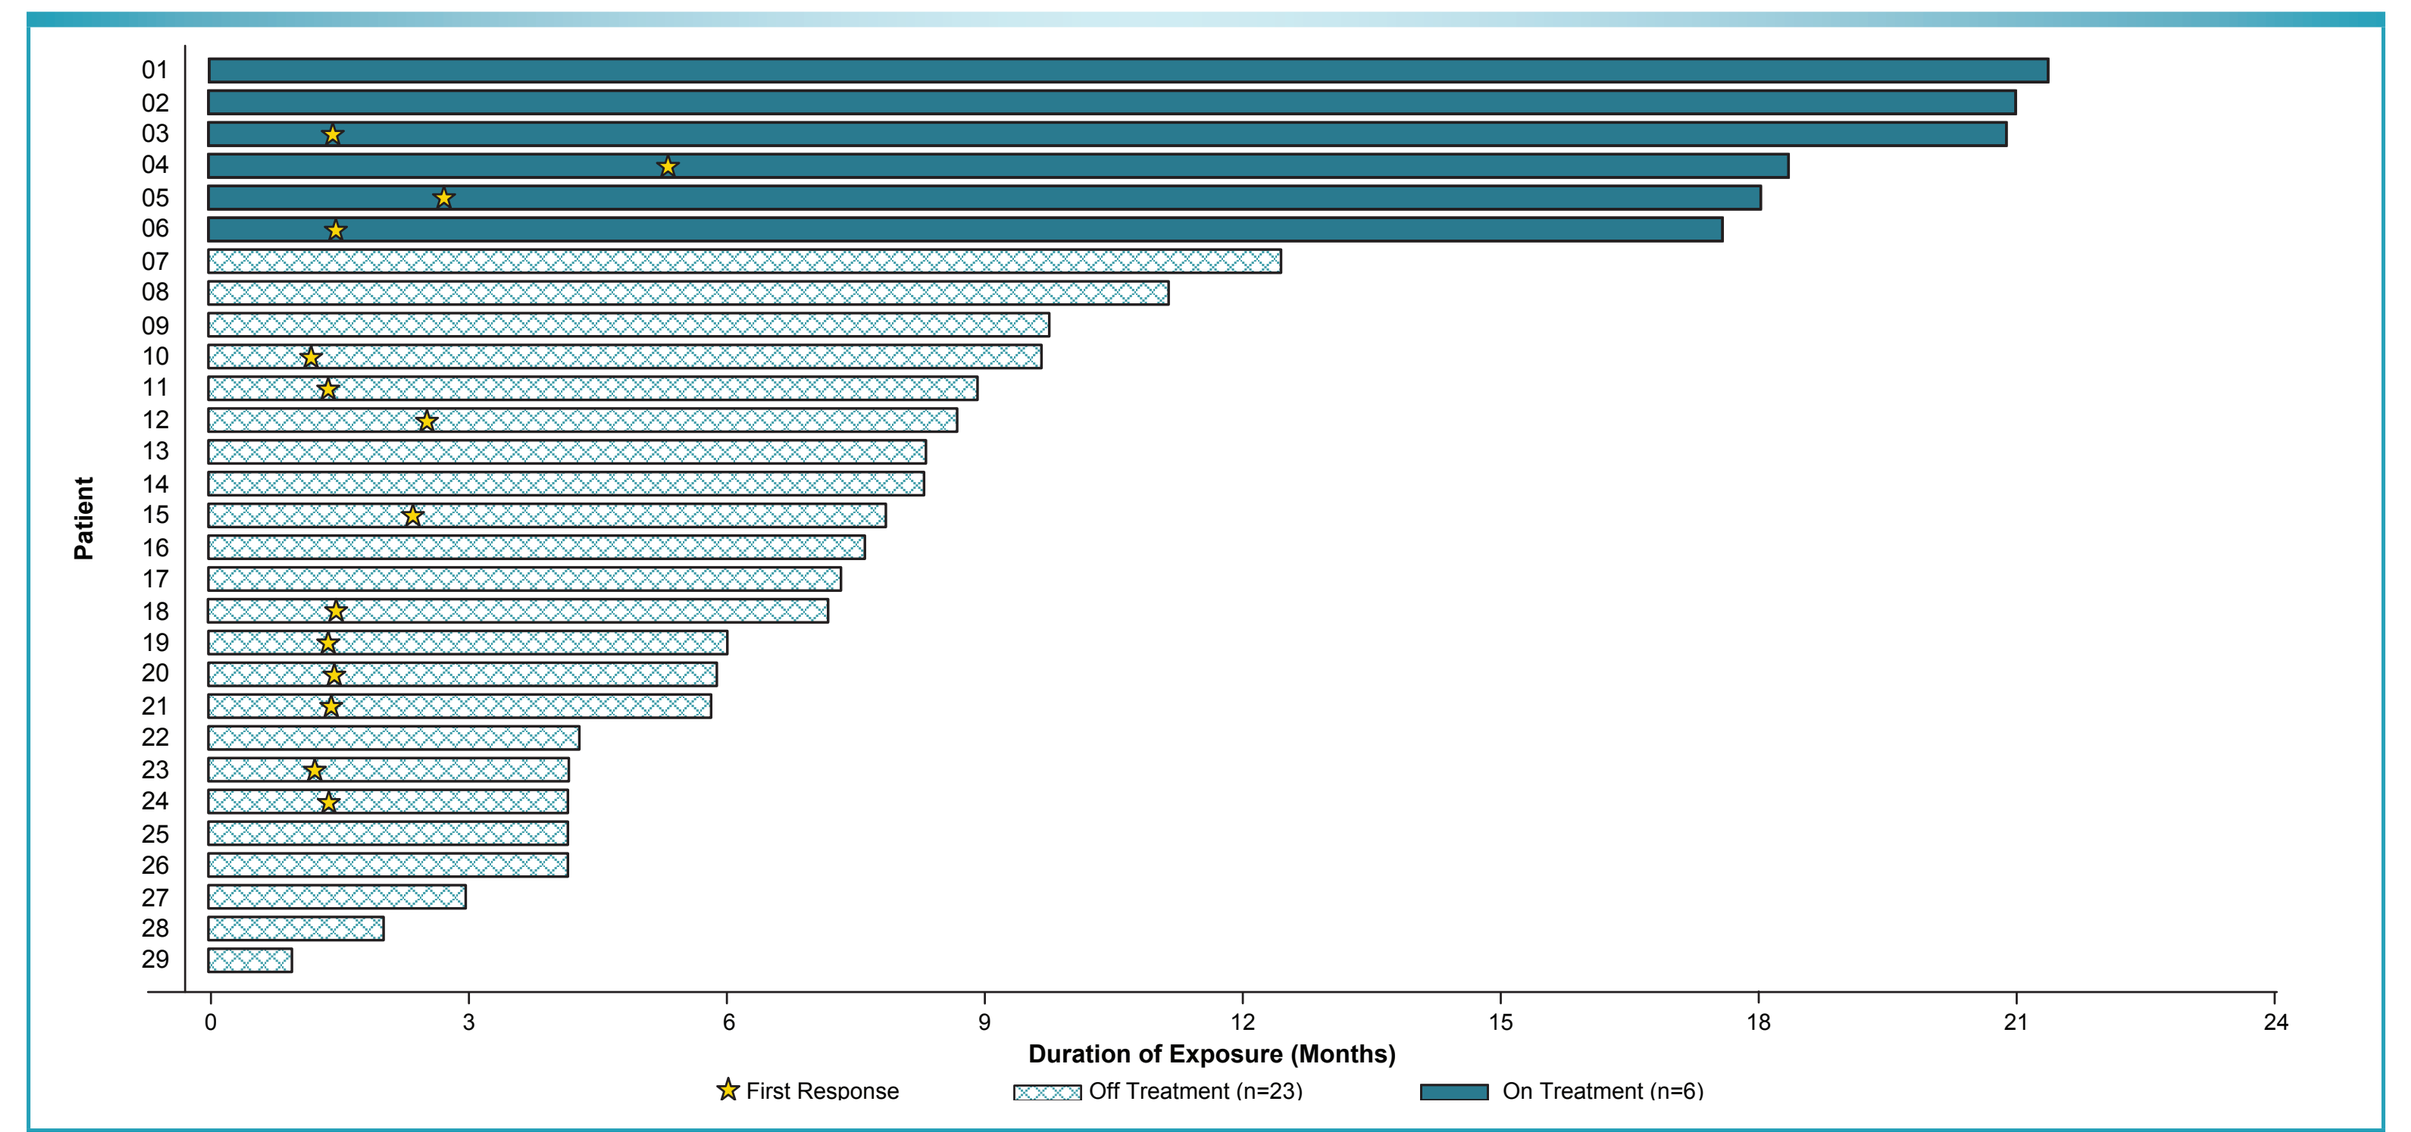

Patient
01
02
03
04
05
06
07
08
09
10
11
12
13
14
15
16
17
18
19
20
21
22
23
24
25
26
27
28
29
0
3
6
9
12
15
18
21
24
Duration of Exposure (Months)
First Response
Off Treatment (n=23)
On Treatment (n=6)

- The median follow-up time for survival was 18.2 months.
- Median PFS was 8.0 months (95% CI, 5.6–9.3 months) per local assessment (Figure 4)

Figure 4. Kaplan-Meier Estimate of Median PFS

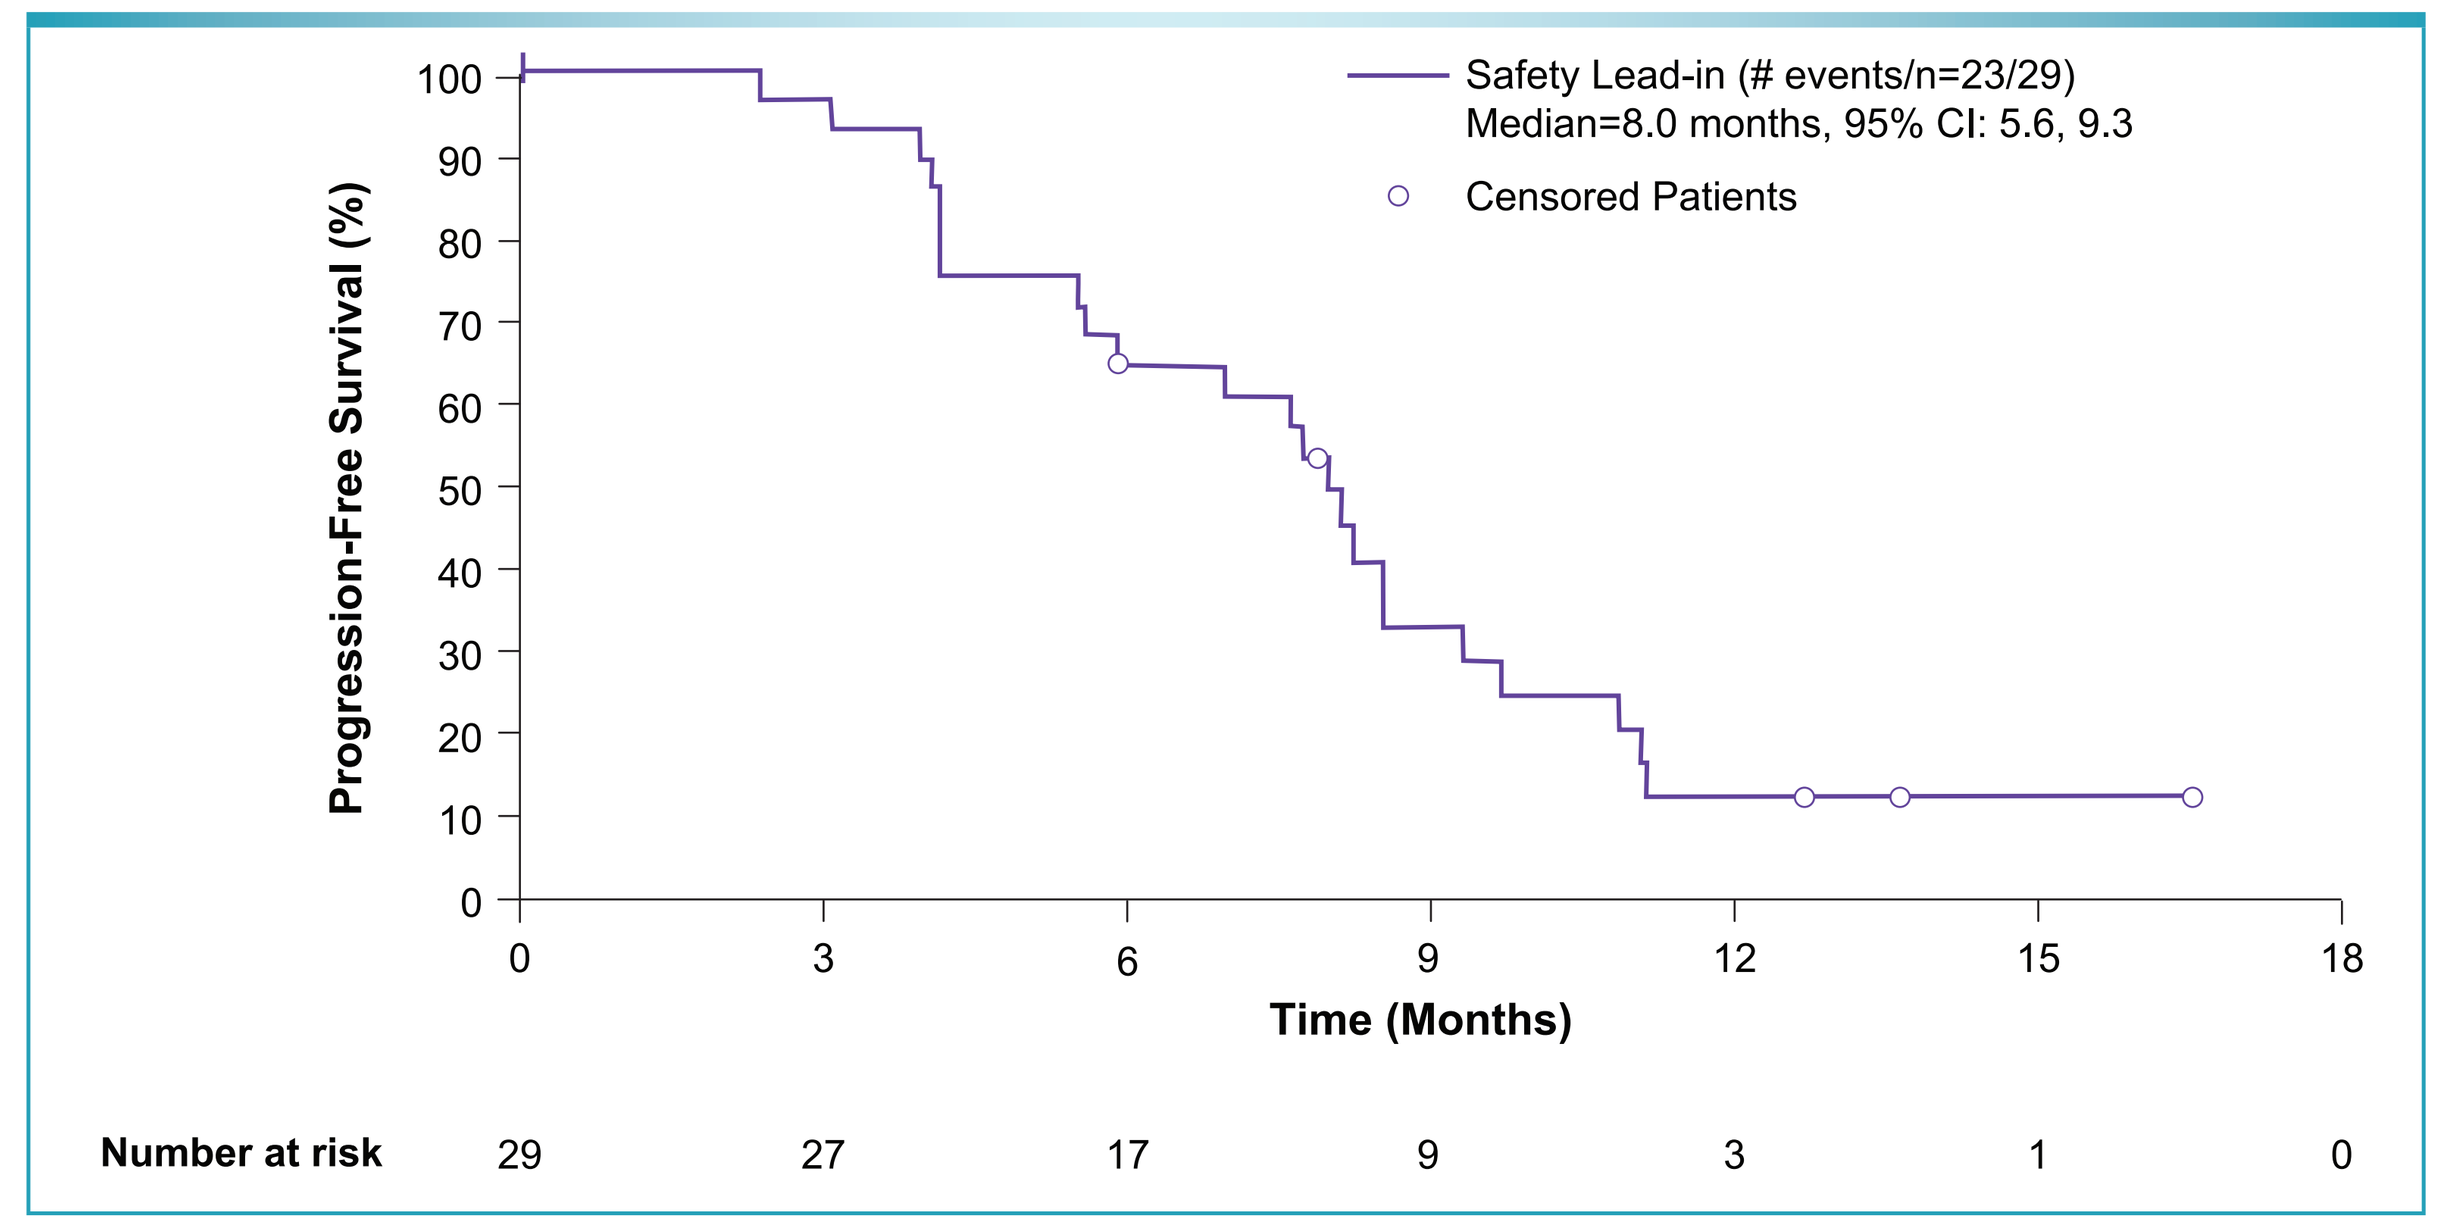

Safety Lead-in (# events/n=23/29)
Median=8.0 months, 95% CI: 5.6, 9.3
Censored Patients
Progression-Free Survival (%)
100
90
80
70
60
50
40
30
20
10
0
0
3
6
9
12
15
18
Time (Months)
Number at risk
29
27
17
9
3
1
0

- The median OS was 15.3 months (95% CI, 9.6 months–NR) (Figure 5)
 - 6-month OS; 86.2% (67.3–94.6)
 - 12-month OS; 62.1% (42.1–76.9)

Figure 5. Kaplan-Meier Estimate of Median OS

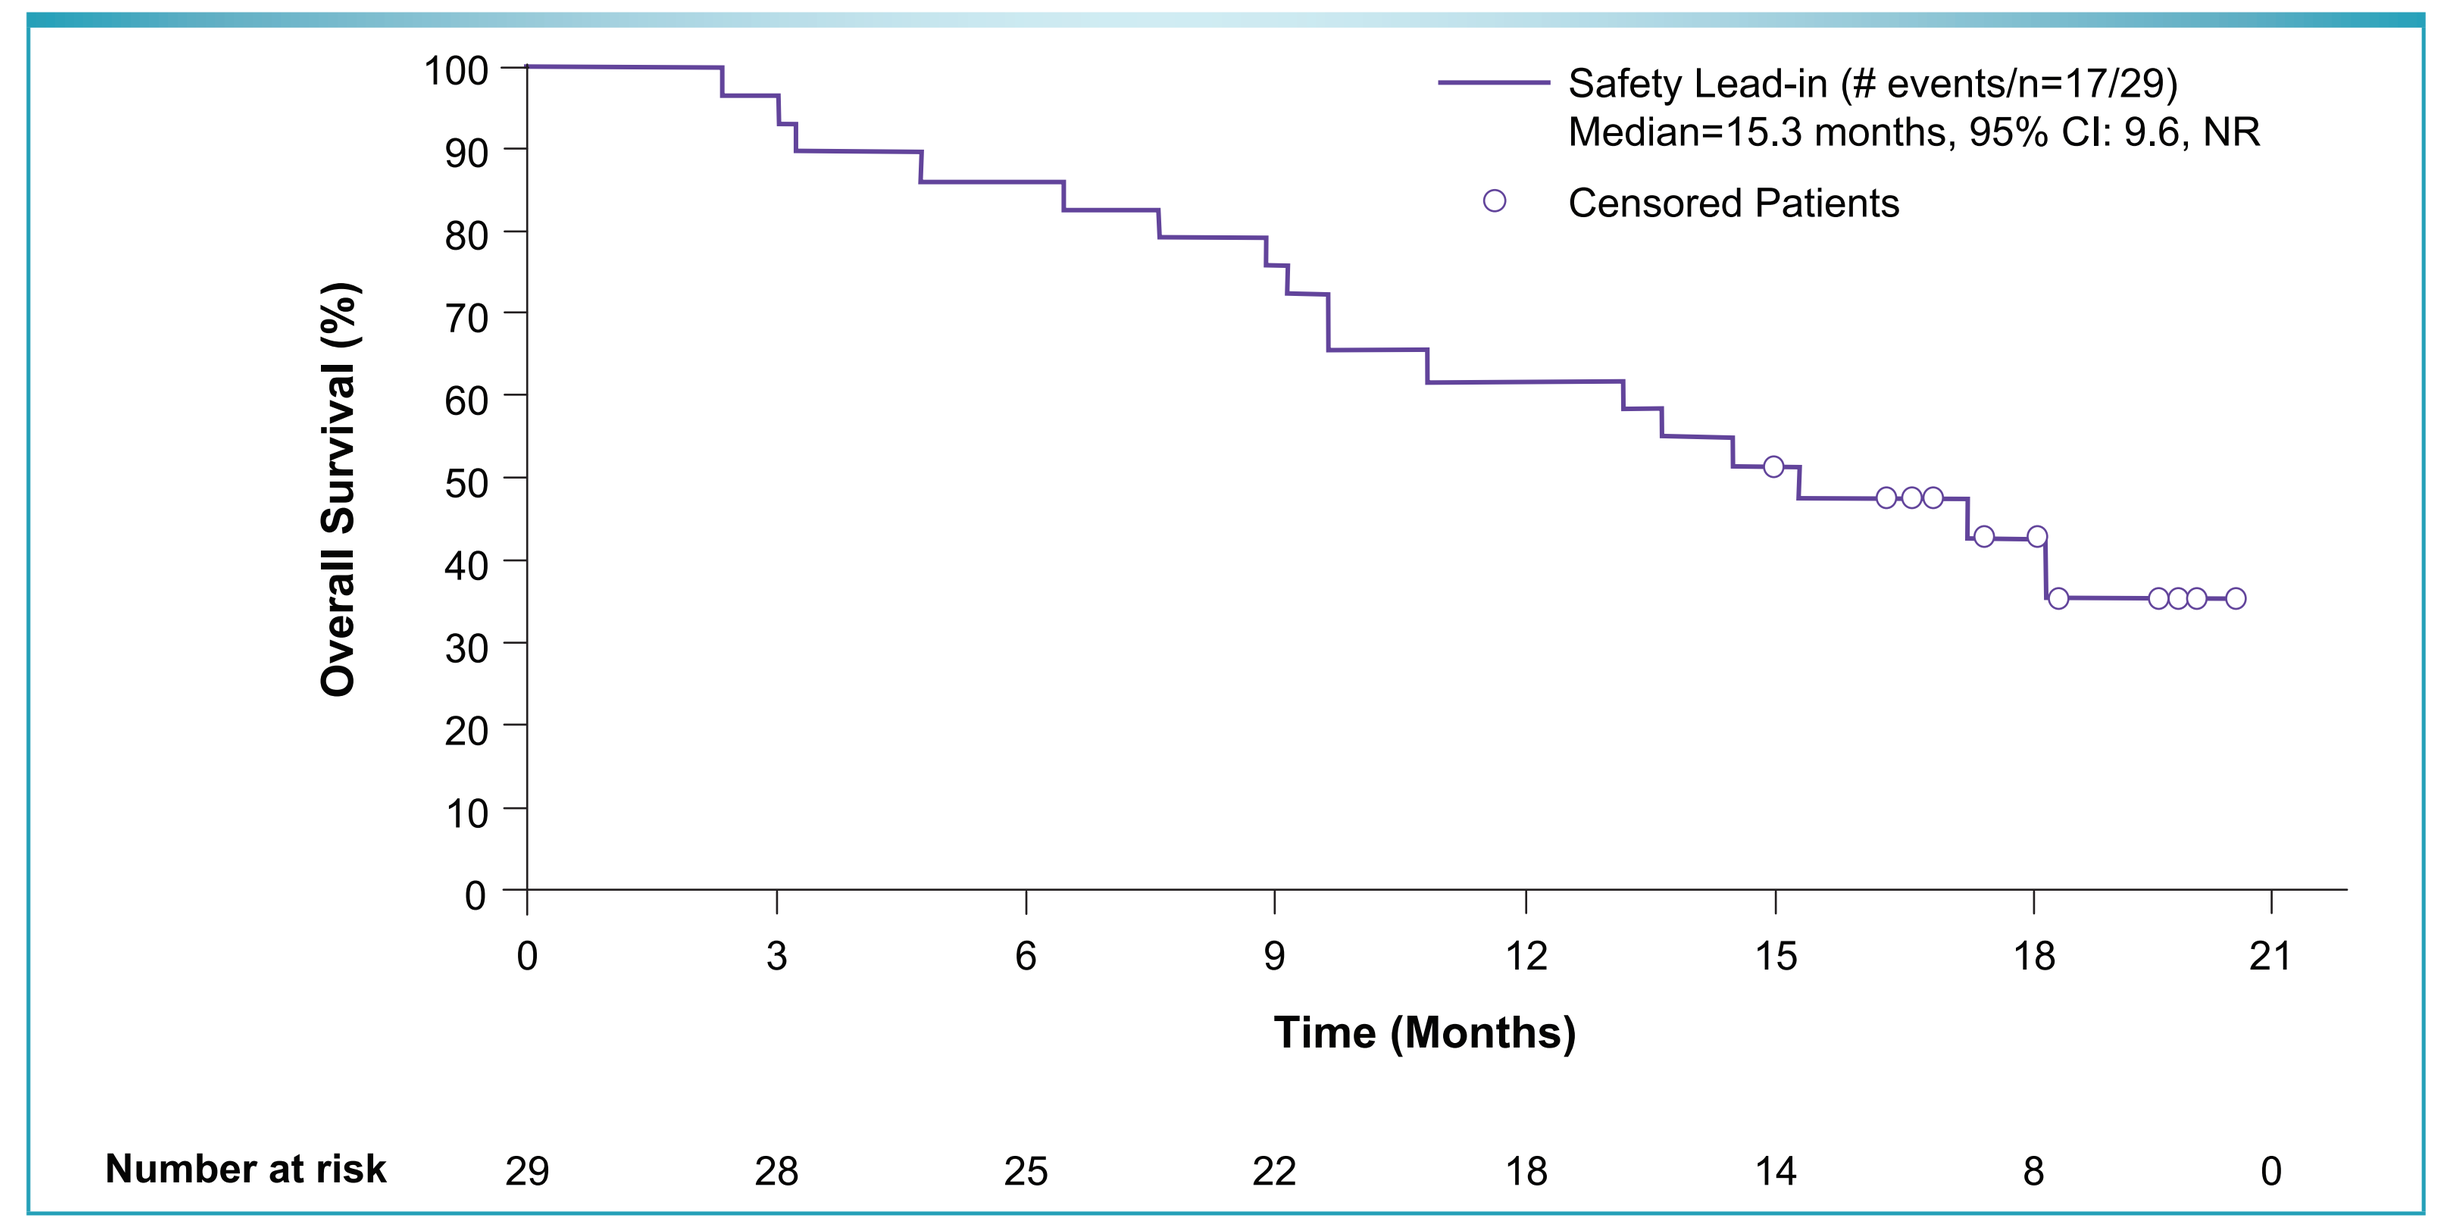

Safety Lead-in (# events/n=17/29)
Median=15.3 months, 95% CI: 9.6, NR
Censored Patients
Overall Survival (%)
100
90
80
70
60
50
40
30
20
10
0
0
3
6
9
12
15
18
21
Time (Months)
Number at risk
29
28
25
22
18
14
8
0

CONCLUSIONS

- ENCO and BINI combined with CETUX has a manageable toxicity profile, with efficacy exceeding historic data in patients with *BRAF* V600–mutant mCRC
- Median OS was 15.3 months (95% CI, 9.6–not reached), with median duration of follow-up of 18.2 (16.6–19.8) months
- Overall response rate by central assessment was consistent with local assessment
- The confirmed ORR and median PFS remain unchanged from the previous report (ORR, 48% [95%CI, 29.4–67.5]; PFS, 8.0 mo [95% CI, 5.6–9.3 mo] - Local Assessment)
 - Historic standards of care have generally demonstrated ORRs <10%, median PFS rates of ≈2 months, and median OS times of 4–6 months
- Common AEs were similar to those previously reported with other *BRAF*, MEK, and EGFR inhibitors and included gastrointestinal and skin toxicities
 - Only one patient discontinued treatment with all drugs due to an adverse event
 - The rate of grade 3/4 skin toxicities continues to be lower than generally observed with CETUX in mCRC
- On the basis of results from the BEACON SLI, the randomized phase 3 portion of the study was initiated and is currently ongoing
- If the data are consistent with the SLI, the results of the phase 3 BEACON CRC study could establish this regimen as a new standard of care for patients in this setting

REFERENCES

1. Loupakis F, et al. *Br J Cancer.* 2009;101:715-721.
2. Tie J, et al. *Int J Cancer.* 2011;128:2075-2084.
3. De Roock W, et al. *Lancet Oncol.* 2010;11:753-762.
4. Morris V, et al. *Clin Colorectal Cancer.* 2014;13:164-171.
5. Giantonio BJ, et al. *J Clin Oncol.* 2007;25:1539-1544.
6. Haller DG, et al. *J Clin Oncol.* 2008;26:4544-4550.
7. Iwamoto S, et al. *Ann Oncol.* 2015;26:1427-1433.
8. Peeters M, et al. *Ann Oncol.* 2014;25:107-116.
9. Sobrero AF, et al. *J Clin Oncol.* 2008;26:2311-2319.
10. Tabernero J, et al. *Lancet Oncol.* 2015;16:499-508.
11. Van Cutsem E, et al. *J Clin Oncol.* 2012;30:3499-3506.
12. Van Cutsem E, et al. *World GI oral presentation.* 2018.
13. Corcoran RB, et al. *Cancer Discov.* 2012;2:227-235.
14. Prahallad A, et al. *Nature.* 2012;483:100-103.
15. Planchard D, et al. *Lancet Oncol.* 2016;17:984-993.
16. Corcoran RB, et al. *Ann Oncol.* 2016;27(suppl 6) [abstract 4550].
17. https://www.clinicaltrials.gov/ct2/show/NCT02928224. Accessed November 15, 2018.
18. Van Cutsem E, et al. *J Clin Oncol.* 2018;36(suppl 4S) [abstract 627].

Acknowledgments

The authors thank the patients, their families, and the sites that participated in this study.

The BEACON CRC trial is being conducted with support from Merck KGaA Darmstadt, Germany (for sites outside of North America), Ono Pharmaceutical, and Pierre Fabre.

Editorial support, funded by Array, was provided by The Medicine Group, LLC., a Division of Bespoke Communications

This study is sponsored by Array BioPharma Inc.

Available at: www.arraybiopharma.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission froam ASCO® and the author of this poster.